SNAP 101: Double-Blind, Placebo/Active-Controlled, Safety and PK/PD Study of INP105 (POD® olanzapine) in Healthy Adults

Shrewsbury SB,[1] Swardstrom M,[1] Satterly KH,[1] Campbell J,[1] Tugiono N,[2] Gillies JD,[3] Hoekman J[1]
[1]Impel NeuroPharma, Seattle, WA, US; [2]Nucleus Network, Melbourne, Australia; [3]Clinical Network Services, Brisbane, Australia

Introduction

A 2008 survey of US Emergency Department staff (ED) found 65% had witnessed physical attacks, 32% of employees reported at least one verbal threat per day and 18% had been assaulted at least once with a weapon[1]. Many were due to acute agitation, but only 6% had a protocol for medication selection and only 40% provided training for staff. During acute agitation episodes (up to 7 million/year in the US[2]), OLZ IM is a favored treatment due to a shorter T_{max} than oral tablets or ODT, but IM administration requires cooperation, is invasive and can be painful. Uncooperative patients require restraint for the administration of OLZ IM, which may be viewed as an assault, reducing trust with medical personnel and increasing the likelihood of staff injuries. When possible, non-injectable forms are preferred during agitation, however, currently approved oral products have slower onset of effect, often requiring labor-intensive observation of the medicated patient until resolved.

Methods

Randomized, double-blind, placebo- and active comparator-controlled, ascending-dose, 2-way, 2-period, incomplete block, crossover Phase 1 trial to compare the safety, tolerability, PK and PD of 3 doses of INP105 (5 mg, 10 mg and 20 mg) with 2 doses of OLZ IM (5 mg and 10 mg) and 1 dose of OLZ-ODT (10 mg) was conducted.

Period 1 was open label; Period 2 was double-blind with at least 14 days between dosing in the 2 periods. Dose escalation was staggered across cohorts to allow a monitoring committee to assess safety and tolerability of INP105 between dose groups.

All subjects were observed as in-patients for at least 72 hours post-dosing of reference therapy and IP. Follow-up occured 4, 5 and 14 days after dosing for each study period.

Methods (contd.)

INP105 is a drug-device combination product consisting of a powder form of OLZ delivered by the Precision Olfactory Delivery (POD) device to the vascular rich upper nasal space for rapid control of agitation in a cooperative or uncooperative patient (with a potentially caregiver-administered dose). For this study a near final formulation of OLZ was administered by the research embodiment of the POD (I231) device. For subsequent studies, INP105 will utilize the final commercial formulation and the commercial POD device. INP105 should provide faster onset of relief compared to oral therapy and be a more accessible dosage form compared to IM therapy without a needle. INP105 may also be suitable for early use by patients who have insight into their condition and recognize early symptoms of agitation before escalating uncontrolled agitation leads to violence and injury to the patient, their caregivers and/or healthcare workers. The objectives of this study in healthy adults were to:

1) Establish safety and tolerability of 3 single ascending doses of INP105
2) Compare PK data for OLZ from 3 INP105 doses with OLZ IM (5 and 10 mg) and orally disintegrating tablets (OLZ-ODT) 10 mg
3) Establish and compare PD effects of INP105 to OLZ IM and OLZ-ODT
4) Explore PK/PD and dose-response relationships for INP105

Conclusion

This study, completed in 2018 (with results expected in December), administering olanzapine to the vascular rich upper nasal space with the novel POD device, should guide further clinical development for a needle-free, easy self or care-giver administered, rapidly effective olanzapine treatment to abort episodes of acute agitation in low-intensity community or ER settings.

Study Design

The original study design of dosing with olanzapine 10 mg IM in 1/3 of all volunteers in Period 1 was modified after clinically significant hypotensive events occurred in the first 2 subjects, leading to the following schedule for the remaining 36 volunteers. At SMC 2, a pre-identified decision was made to dose Cohort 3 with INP105 15 mg, rather than the planned 20 mg. PD effects were assessed using VAS, ACES and DSST (and Vital Signs) for 120 hrs after each dosing.

Cohort 1 (visits):

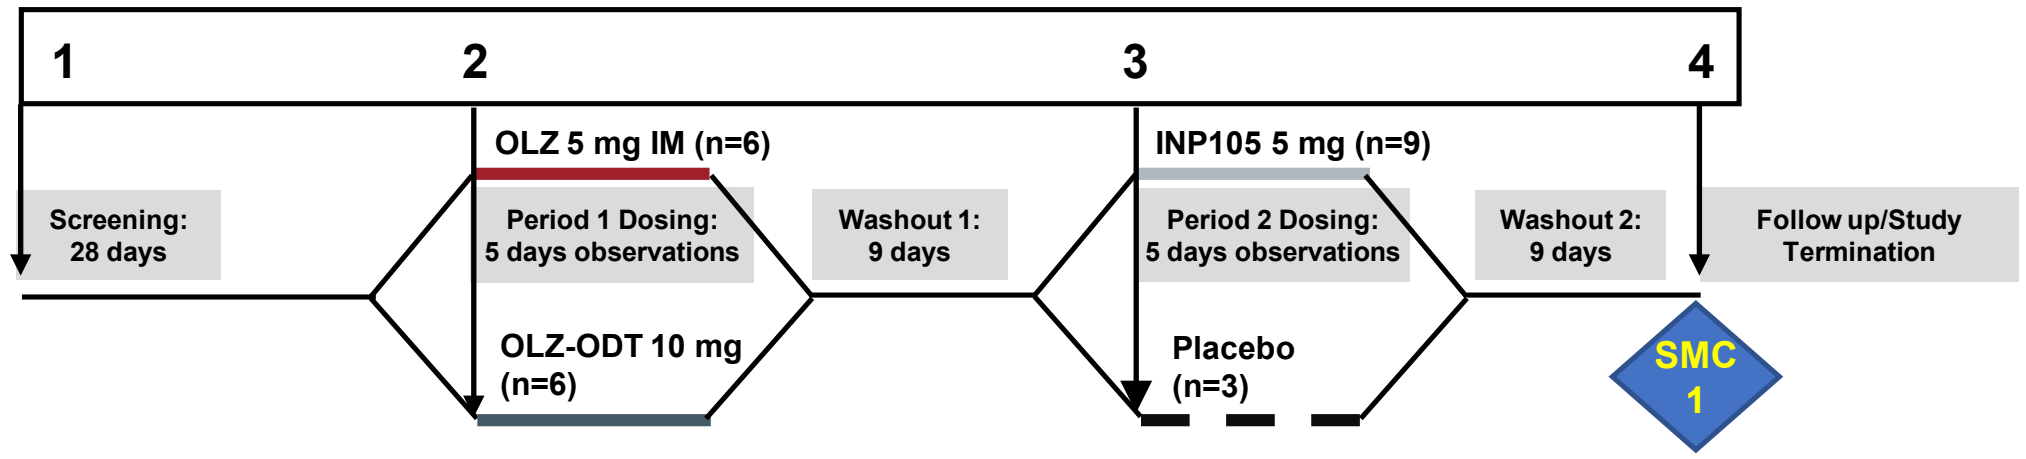
1
2
3
4
OLZ 5 mg IM (n=6)
INP105 5 mg (n=9)
Screening: 28 days
Period 1 Dosing: 5 days observations
Washout 1: 9 days
Period 2 Dosing: 5 days observations
Washout 2: 9 days
Follow up/Study Termination
OLZ-ODT 10 mg (n=6)
Placebo (n=3)
SMC 1

Cohort 2 (visits):

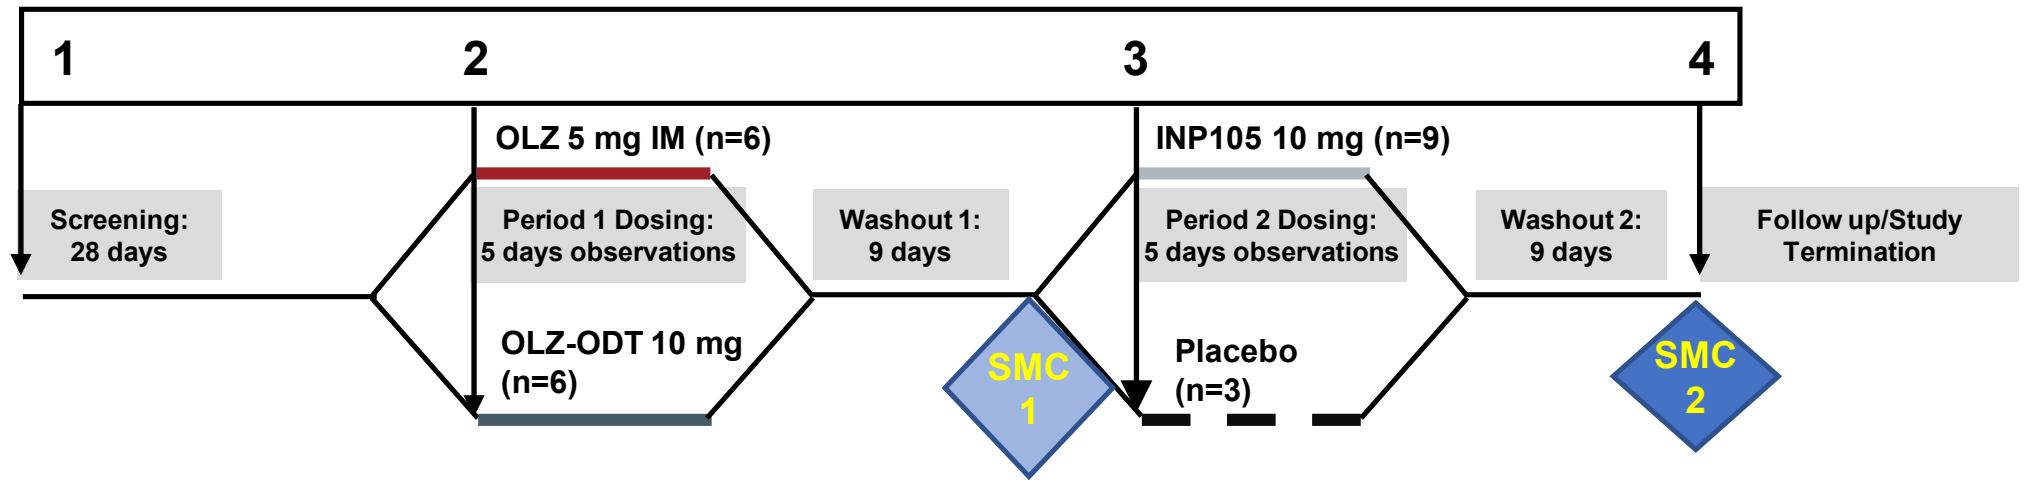
1
2
3
4
OLZ 5 mg IM (n=6)
INP105 10 mg (n=9)
Screening: 28 days
Period 1 Dosing: 5 days observations
Washout 1: 9 days
Period 2 Dosing: 5 days observations
Washout 2: 9 days
Follow up/Study Termination
OLZ-ODT 10 mg (n=6)
SMC 1
Placebo (n=3)
SMC 2

Cohort 3 (visits):

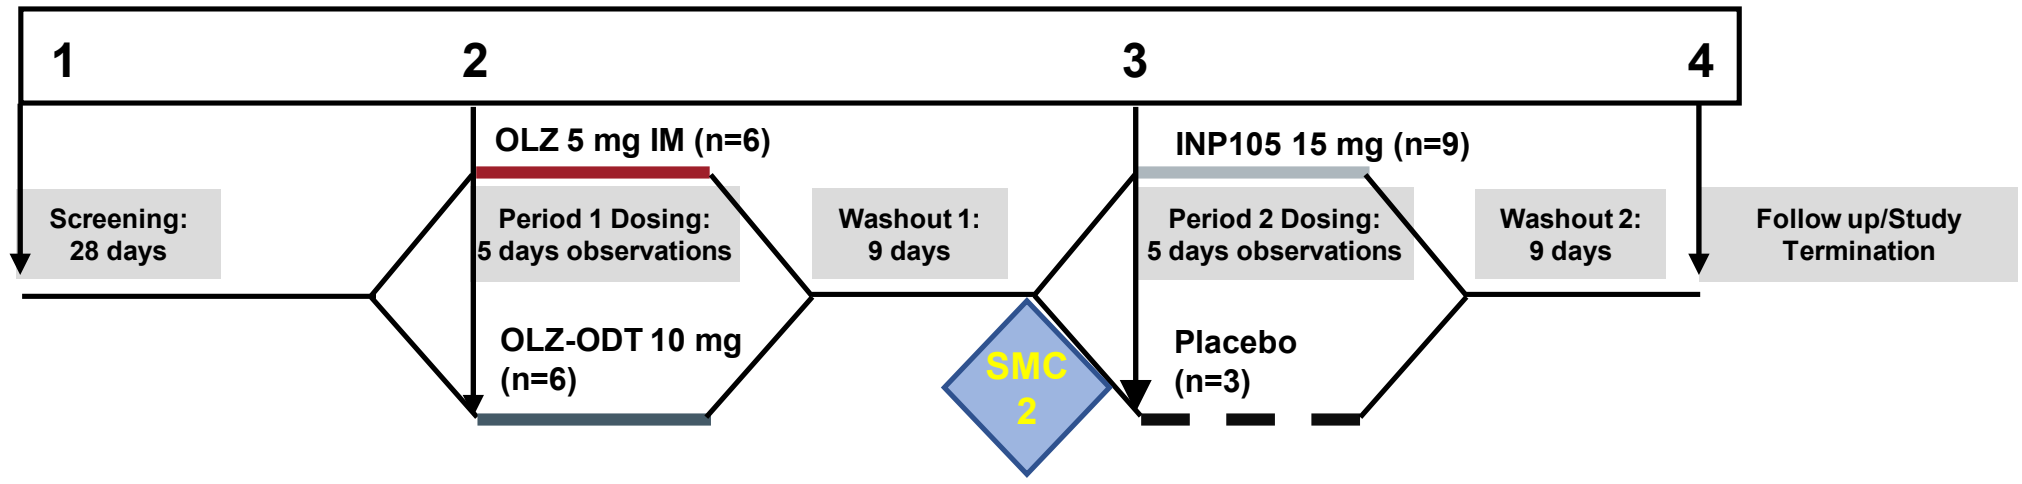
1
2
3
4
OLZ 5 mg IM (n=6)
INP105 15 mg (n=9)
Screening: 28 days
Period 1 Dosing: 5 days observations
Washout 1: 9 days
Period 2 Dosing: 5 days observations
Washout 2: 9 days
Follow up/Study Termination
OLZ-ODT 10 mg (n=6)
SMC 2
Placebo (n=3)

References: [1] Kansagra SM, Rao SR, Sullivan AF, et al. A survey of workplace violence across 65 U.S. emergency departments. Acad Emerg Med. 2008;15(12):1268-74.
[2] Sachs GS. A review of agitation in mental illness: burden of illness and underlying pathology. J Clin Psychiatry. 2006;67(10):5-12.

Impel NEUROPHARMA