Does Dihydroergotamine Treat the "Whole Migraine"?

Sheena K. Aurora, MD,[1*] Sutapa Ray, PhD,[1] Kelsey Satterly, PhD,[1] Stephen B. Shrewsbury, MB ChB,[1] John Hoekman, PhD[1]

[1]Impel NeuroPharma, Seattle, WA

*Presenting Author

Introduction

The Whole Migraine: Phases of the Migraine Cycle

- Migraine is a complex and multifaceted disorder with 5 distinct phases; therefore, it is important to consider treating the whole migraine (**Figure 1**)[1]

Figure 1. The Five Phases of the Migraine Cycle Include the Premonitory, Aura, Headache, Postdrome, and Interictal Phases[1-4]

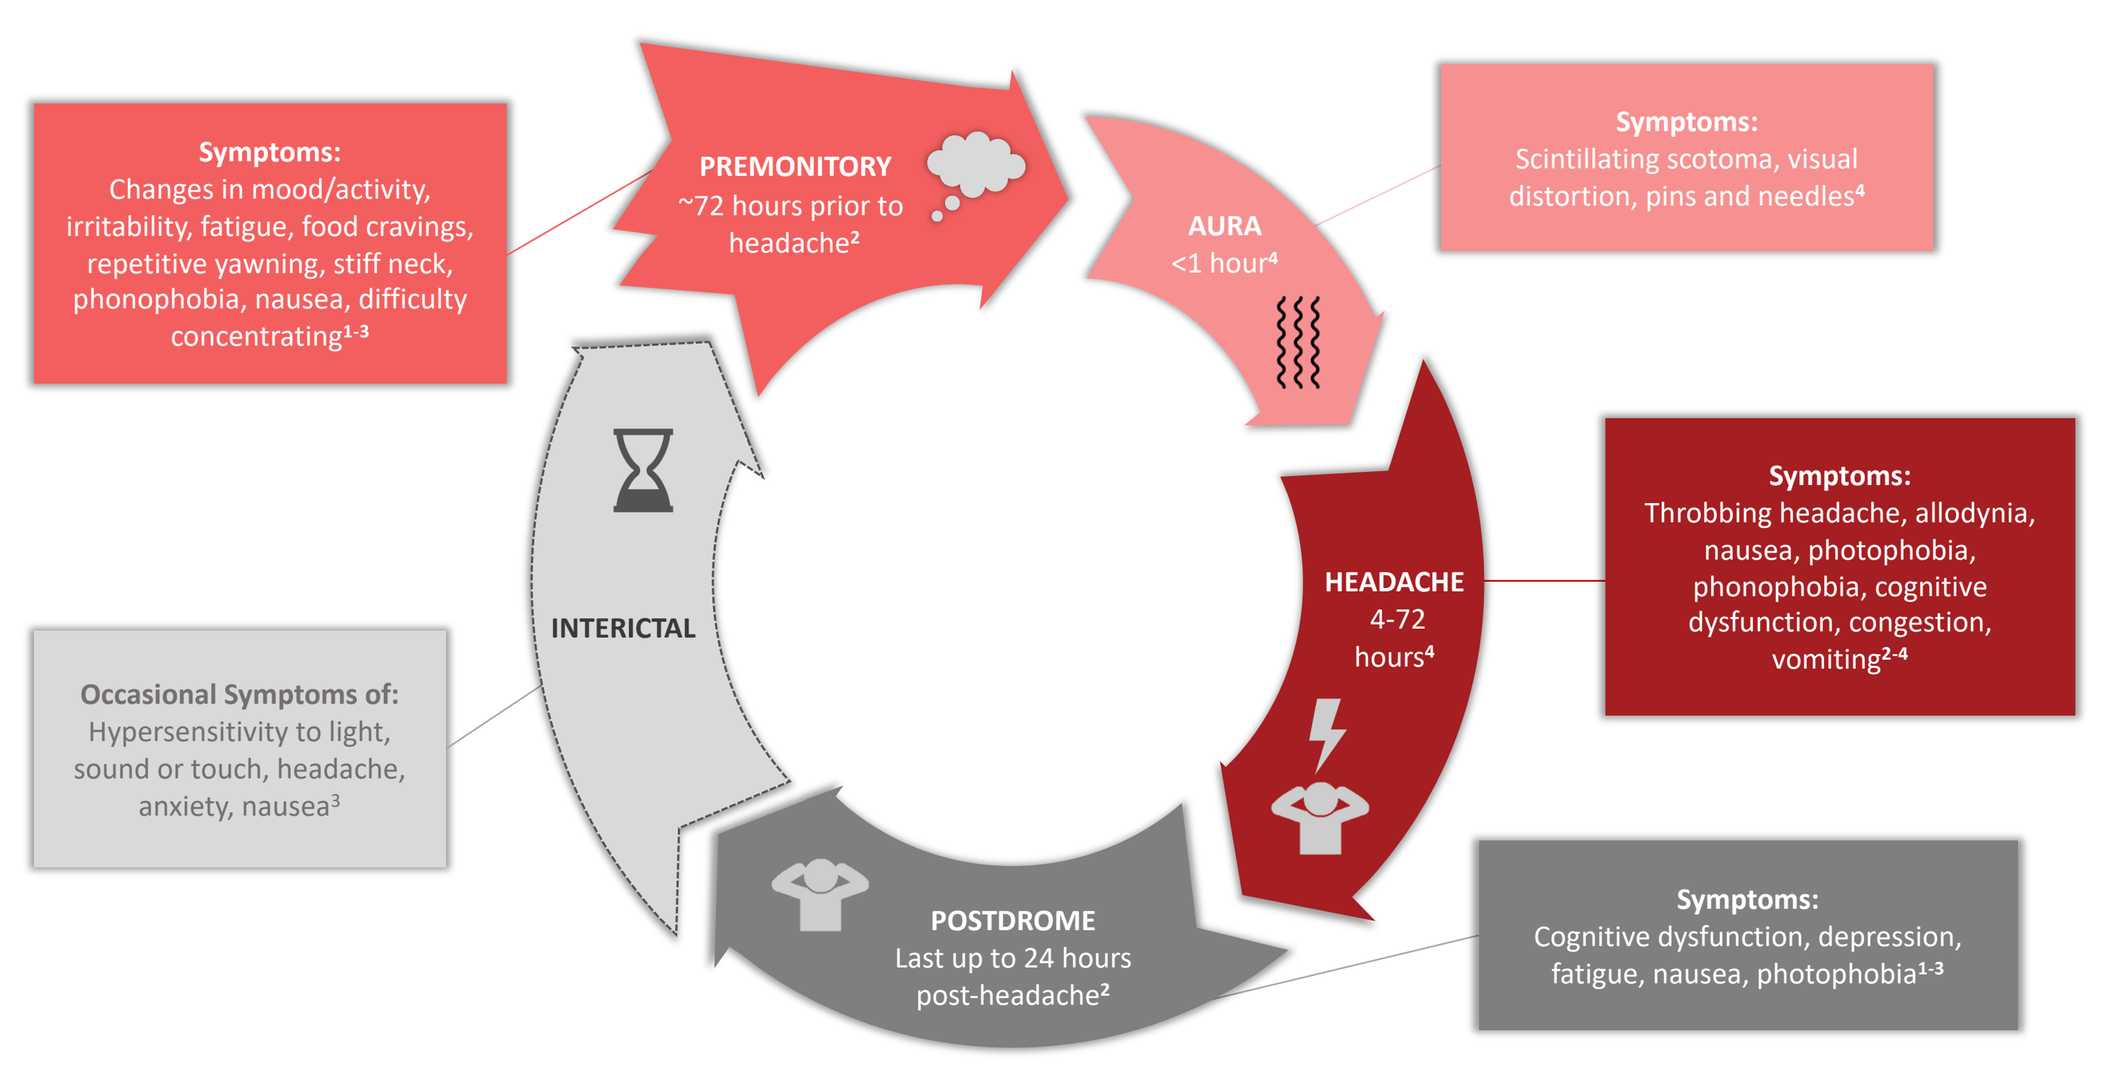
Symptoms: Changes in mood/activity, irritability, fatigue, food cravings, repetitive yawning, stiff neck, phonophobia, nausea, difficulty concentrating[1-3]
PREMONITORY ~72 hours prior to headache[2]
AURA <1 hour[4]
Symptoms: Scintillating scotoma, visual distortion, pins and needles[4]
HEADACHE 4-72 hours[4]
Symptoms: Throbbing headache, allodynia, nausea, photophobia, phonophobia, cognitive dysfunction, congestion, vomiting[2-4]
POSTDROME Last up to 24 hours post-headache[2]
Symptoms: Cognitive dysfunction, depression, fatigue, nausea, photophobia[1-3]
INTERICTAL
Occasional Symptoms of: Hypersensitivity to light, sound or touch, headache, anxiety, nausea[3]

Current Migraine Therapies and Limitations

- One of the primary goals for the treatment of migraine attacks includes the rapid relief of pain and associated symptoms[5]
- Most migraine therapies target a very narrow set of receptors focused mainly on headache pain
 - Triptans are commonly used as acute medications and are 5-hydroxytryptamine (5-HT)$_{1B/1D}$ receptor agonists with some affinity for the 5-HT$_{1F}$ receptor subtype[6]
 - Novel emerging acute and preventive therapies include ditans (5-HT$_{1F}$ receptor agonists), gepants (calcitonin gene-related peptide [CGRP] receptor antagonists), and anti-CGRP monoclonal antibodies[6]
- Because migraine encompasses a spectrum of symptoms, this narrowly targeted receptor profile for most migraine therapies does not allow patients to achieve a holistic relief from migraine, and patients often discontinue treatment due to a lack of consistency, headache recurrence, and accompanying nausea[1,5]

Dihydroergotamine (DHE)

- DHE has a long, established history as an effective migraine therapy and is well-regarded by physicians because of its[7,8]:
 - Rapid onset[9,10]
 - Efficacy against a full range of acute symptoms of migraine, including pain, photophobia, and phonophobia[11]
 - Efficacy irrespective of the time of treatment (**Table 1**)[12]
- DHE is effective in patients with difficult-to-treat migraine, such as those who[8]:
 - Have status migrainosus[7]
 - Wake with migraine[13]
 - Are triptan resistant[13]
 - Have menstrual migraine[14]
 - Have allodynia[11,13]
 - Have severe or prolonged migraine[9,11,13]
- There is also minimal risk of medication overuse with DHE use[7]

Table 1. The Pharmacokinetics of Different Routes of Administration of DHE

DHE Pharmacokinetics				
DHE Route of Administration	**Dose (mg)**	**T_{max} (Minutes)**	**$AUC_{0\text{-}2hours}$ (h*pg/mL)**	**Absolute Bioavailability***
Intravenous[7,15]	1	1-6	3019	100%
Intramuscular[7,16]	1	15-24	4791	100%
Subcutaneuous[8,17]	1	20-40	N/A	100%
Nasal spray[7]	2	48-60	428.7-1316	32%[†]
POD®[7,18]	1.45	30	1595	59%

AUC, area under the curve; DHE, dihydroergotamine; N/A, not available; POD, precision olfactory delivery.

*Absolute bioavailability is defined as the amount of drug from a formulation that reaches systemic circulation relative to an intravenous dose.

[†]Nasal spray refers to Migranal®, which has a variable bioavailability; 32% according to the prescribing information and 13-101% according to the Summary Basis of Approval.[19]

Methods

This is a review of the comparative pharmacology of acute treatments for migraine performed by conducting a literature review of the pharmacology and biological activity of new and existing migraine-specific treatments

Objectives

- To compare receptor pharmacology of acute treatments for migraine in published literature
- To examine the pharmacology of acute treatments for migraine in the context of the migraine cycle to provide insight into the total migraine benefits of treating with a broad receptor binding agent, such as DHE, versus the limitations of treating with narrowly targeted therapies
- To find a possible explanation for why DHE has such a high response rate even in the most difficult-to-treat migraine

Results

Table 2. Compared to Other Acute Migraine Therapies, DHE Displays a Broad Range of Pharmacological Activity at Serotonergic (5-HT), Adrenergic (α), and Dopaminergic (D) Receptor Subtypes

■ Potent agonist ■ Partial agonist ■ Antagonist ■ Partial Antagonist

Receptor	IV DHE/ DHE[20-22]	Inhaled DHE[20]	Triptans[23]	Ditans[23]	Gepants[6]	Receptor Distribution[24-28]
5-HT$_{1A}$	Potent agonist	Potent agonist				raphe nuclei, hippocampus
5-HT$_{1B}$	Potent agonist	Potent agonist	Potent agonist			substantia nigra, globus pallidus, basal ganglia
5-HT$_{1D}$	Potent agonist	Potent agonist	Potent agonist	Partial agonist		brain
5-HT$_{1E}$				Partial agonist		cortex, putamen
5-HT$_{1F}$	Partial agonist		Potent agonist	Potent agonist		cortex, hippocampus
5-HT$_{2A}$	Antagonist	Partial Antagonist				platelets, smooth muscle, cerebral cortex
5-HT$_{2B}$	Partial agonist					stomach fundus
5-HT$_{2C}$	Partial agonist					choroid, hippocampus, substantia nigra
5-HT$_3$						area postrema, sensory and enteric nerves
5-HT$_4$						CNS and myenteric neurons, smooth muscle
5-HT$_5$						brain
5-HT$_6$						brain
5-HT$_7$	Antagonist					brain
α$_{1a}$	Antagonist	Partial Antagonist				postsynaptic effector cells (smooth muscle), brain, heart, liver, spleen
α$_{1b}$						
α$_{2a}$	Antagonist					presynaptic adrenergic nerve terminals, platelets, lipocytes, smooth muscle
α$_{2b}$	Antagonist	Partial Antagonist				
α$_{2c}$						
β$_1$						postsynaptic effector cells (heart, lipocytes, brain), presynaptic adrenergic and cholinergic nerve terminals, kidney, eye
β$_2$						postsynaptic effector cells (smooth muscle, cardiac muscle)
β$_3$						postsynaptic effector cells, especially lipocytes; heart
D$_2$	Antagonist	Antagonist				brain, vascular, heart, sympathetic ganglia
D$_3$						brain, vascular, kidney
D$_4$						brain, heart, kidney
CGRP					Antagonist	PNS, CNS, periphery
EphrinB2	Antagonist					brain, heart, lung, kidney, placenta, pancreas, liver and skeletal muscle

CGRP, calcitonin gene-related peptide; CNS, central nervous system; DHE, dihydroergotamine; IV, intravenous; PNS, peripheral nervous system.

Note: Functional receptor binding data presented in this table is not a direct comparison, but a review of available, published data on functional receptor binding for acute migraine drug therapies. Inhaled DHE meslyate is MAP0004.

Figure 2. Hypothetical Model for How DHE Targets the Whole Migraine

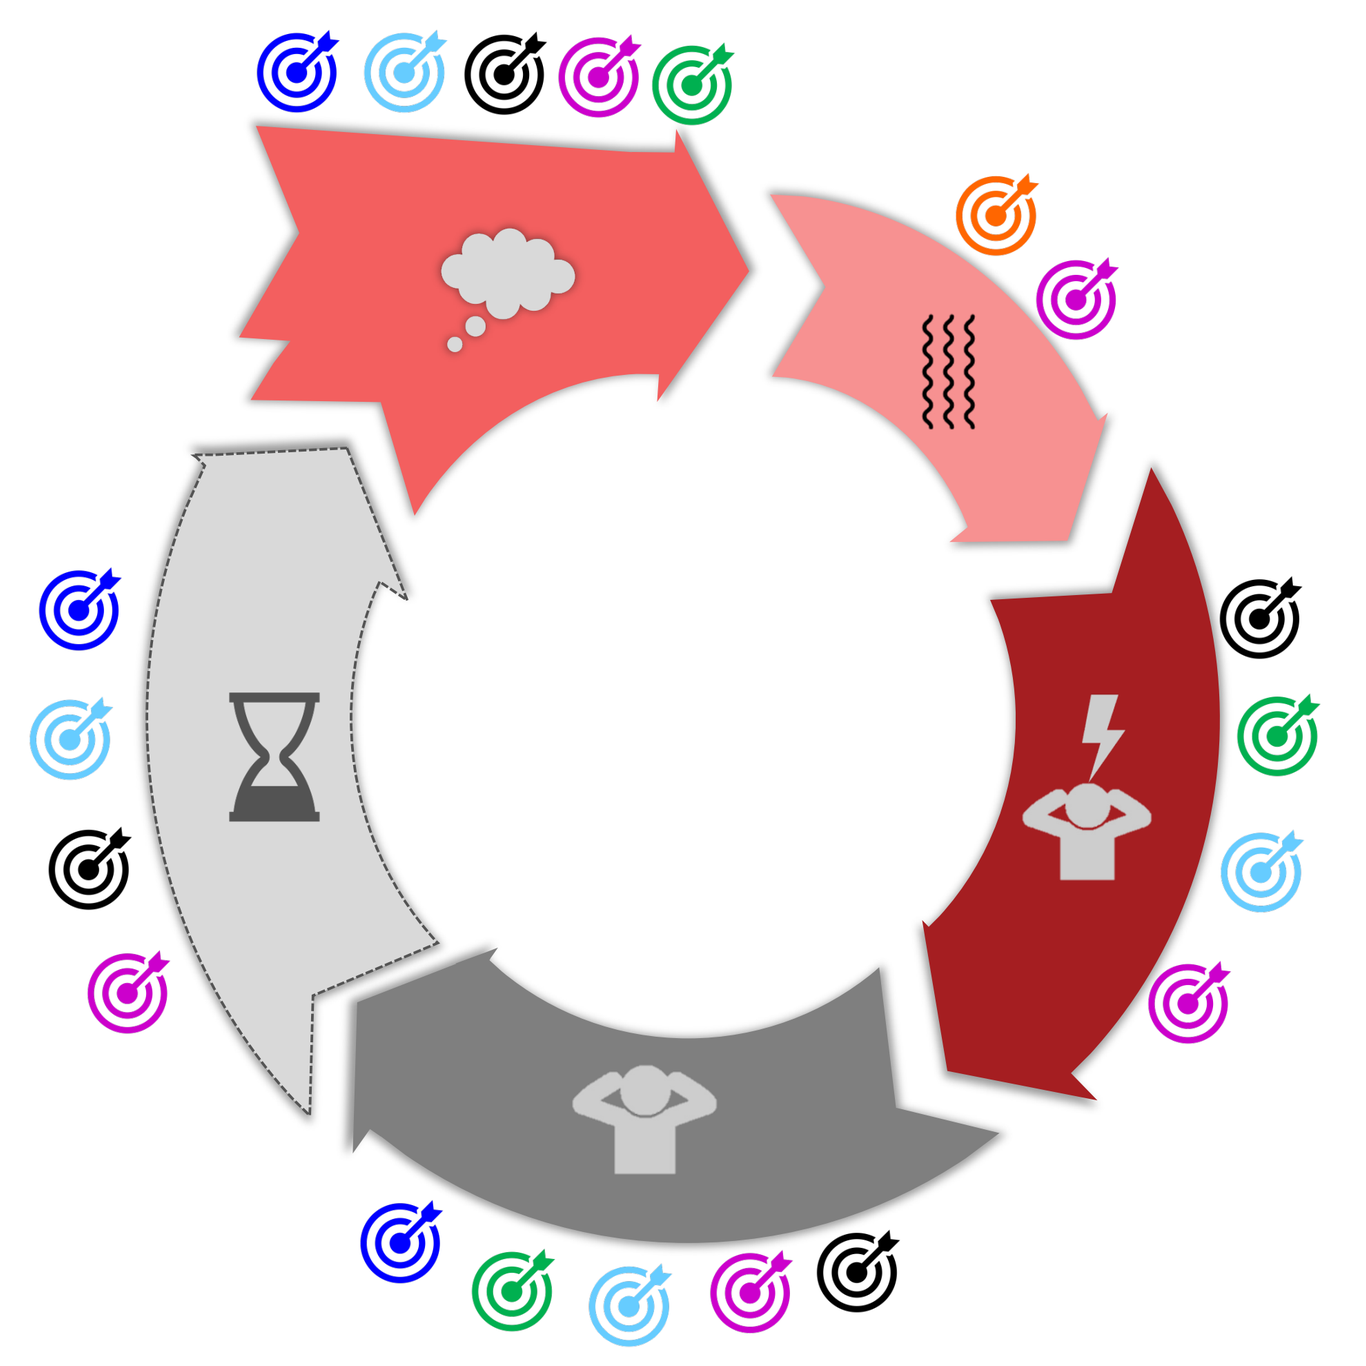

KEY

- DHE can alleviate headache pain symptoms via several mechanisms
 - Constricts pain-producing intracranial extracerebral blood vessels via 5-HT$_{1B}$ receptors[16]
 - Inhibits trigeminal neurotransmission via activation of peripheral and central 5-HT$_{1D}$ receptors[16]
 - Effective in patients with allodynia[11]
 - Agonist of 5-HT$_{1A}$ receptors, which may have a beneficial role in migraine prophylaxis, which may lengthen the interictal period[20]
 - Central effects of DHE may reverse central sensitization[8]
 - Antagonist of 5-HT$_{2A}$ receptors, a pronociceptive receptor that can increase headache attacks[29]
 - It has been suggested that antimigraine effects of DHE involve repressing CGRP release via activation of α2-adrenoceptors[30]
- May confer anti-nausea activity via D$_2$ receptor antagonism[20]
- Is effective for acute treatment of migraine with aura[12]
- Has 5-HT$_{2A}$ receptor antagonism, which is involved in mood disorders[30]
- Exerts D$_2$ antagonism, and a dopamine imbalance during migraine attacks may contribute to pain, discomfort, increased sensory sensitivity, and aversive reactions to environmental stimuli[32]
- Binds to the ephrinB2 receptor which has been implicated in memory, and DHE has been shown to improve memory problems in post-concussive syndrome[33,34]

Conclusion

- Unlike other migraine therapeutics, DHE interacts with several receptor families and subtypes, which include serotonergic, adrenergic, and dopaminergic subtypes (**Table 2**)
- DHE also slowly dissociates from 5-HT$_{1B/1D}$ receptor sites, which may explain why DHE has sustained anti-migraine effects[35]
- DHE is able to exert a greater influence than single receptor agonists/antagonists over the pathophysiology of the migraine cycle due to its widespread pharmacological activity (**Figure 2**)
- DHE administered at consistent doses and optimal plasma concentrations not only maximizes therapeutic gain, but also improves safety and tolerability[7,20]
- Advances in DHE delivery systems will further address these issues[7,20]

References

1. Charles A. *Headache.* 2013;53(2):413-419.
2. Goadsby PJ, et al. *Physiol Rev.* 2017;97(2):553-622.
3. Linde M. *Acta Neurol Scand.* 2006;114(2):71-83.
4. Headache Classification Committee of the International Headache Society. *Cephalalgia.* 2018;38(1):1-211.
5. Lipton RB, et al. *Headache.* 2019;59(10):1762-1772.
6. Do TP, et al. *J Headache Pain.* 2019;20(1):37.
7. Silberstein SD, et al. *Headache.* 2020;60(1):40-57.
8. Baron EP, Tepper SJ. *Future Neuro.* 2011;6(3):327-333.
9. Saper JR, et al. *Headache.* 2006;46(Suppl 4):S212-220.
10. Kori SH, et al. *Ann Neurol.* 2010;68(S14).M-41.
11. Tepper SJ, et al. *Headache.* 2012;52(1):37-47.
12. Tepper SJ, et al. *Mayo Clin Proc.* 2011;86(10):948-955.
13. Kori SH, et al. *Ann Neurol.* 2010;68(S14).M-48.
14. Aurora S, et al. *J Headache Pain.* 2013;14(Suppl 1):P143.
15. Saper JR, Silberstein S. *Headache.* 2006;46(Suppl 4):S171-S181.
16. Silberstein SD, McCrory DC. *Headache.* 2003;43(2):144-166.
17. Tfelt-Hansen PC. *Cephalalgia.* 2013;33(13):1122-1131.
18. Shrewsbury SB, et al. *Headache.* 2019;59(3):394-409.
19. Satterly KH, et al. *Headache.* 2019;59:1-208.#P134.
20. Cook RO, et al. *Headache.* 2009;49(10):1423-1434.
21. Guzman M, et al. *Neurology.* 2020;94(S15). #4425.
22. Suzuki K, et al. *FEBS Open Bio.* 2016;6(5):461-468.
23. Rubio-Beltrán E, et al. *Br J Pharmacol.* 2019;176(24):4681-4695.
24. Katzung BG, et al. *Basic & Clinical Pharmacology.* New York: McGraw-Hill Medical, 2012.
25. Perez-Lloret S, et al. *Expert Opin Drug Saf.* 2014;13(3):351-360.
26. Schwinn DA. *Adv Pharmacol.* 1994;31:333-341.
27. Uniprot. https://www.uniprot.org/uniprot/P29323 (Accessed May 14, 2020).
28. Deen M, et al. *J Headache Pain.* 2017;18(1):96.
29. Srikiatkhachorn A, et al. *Headache.* 2008;42:566-574.
30. Masterson CG, Durham PL. *Headache.* 2010;50(9):1424-1439.
31. Marek GJ, et al. *Neuropsychopharmacology.* 2003;28(2):402-412.
32. DaSilva AF, et al. *Neurology.* 2017;88(17):1634-1641.
33. Suzuki K, et al. *FEBS Open Bio.* 2016;6(5):461-468.
34. McBeath JG, Nanda A. *Headache.* 1994;34(3):148-151.
35. Kori S, et al. *J Headache Pain.* 2013;14(Suppl 1):P75.

Disclosures and Acknowledgments

All authors are full-time employees and stockholders of Impel NeuroPharma. This research was sponsored by Impel NeuroPharma. Editorial support was provided by IMPRINT Science, and funded by Impel NeuroPharma.

Impel NEUROPHARMA®

Presented at the 2020 DHCREF Virtual Meeting: The New Paradigm of Treatment for the Headache Patient