Treatment Consistency Across Multiple Migraine Attacks: Results From the Phase 3 Open-Label STOP 301 Study

Richard B. Lipton, MD[1]*; Barbara L. Nye, MD[2]; Joe Hirman, PhD[3]; Stephen B. Shrewsbury, MB ChB[4]; Sheena K. Aurora, MD[4]

[1]Albert Einstein College of Medicine and Montefiore Headache Center, Bronx, NY, USA; [2]Department of Neurology, Dartmouth-Hitchcock Medical Center, Lebanon, NH, USA; [3]Pacific Northwest Statistical Consulting, Woodinville, WA, USA; [4]Impel NeuroPharma, Seattle, WA, USA

*Presenting author

Introduction

- Consistency of response is an important treatment attribute as individuals with migraine treat multiple migraine attacks (MAs)[1,2]
- Insufficient treatment response due to a lack of consistency may lead to more disability, and, hence, have a negative impact on quality of life[2-4]
- Individuals with migraine may experience gastrointestinal (GI) comorbidities and/or GI dysmotility both during and outside an MA, which may contribute to an inconsistent response, and is a particular concern with oral therapies[5-9]
- Historically, intravenous (IV) administration of dihydroergotamine mesylate (DHE) has been an effective acute treatment for migraine that provides consistent relief, but there is a need to deliver DHE rapidly and reliably without an injection[9-12]
- Despite its well-known efficacy, non-IV use of DHE is currently limited because of variable absorption associated with current nasal products that target the lower nasal space, lack of absorption when administered orally, and operational difficulties of parenteral administration[12]
- INP104 is an investigational, novel drug-device product that targets delivery of DHE to the upper nasal space using Precision Olfactory Delivery (POD®) technology[12]
- It is hypothesized that INP104 could provide a highly consistent headache response because:
 - In a Phase 1 study, INP104 demonstrated IV DHE-like plasma levels and more consistent delivery of DHE than MIGRANAL® (Bausch Health Companies, Inc. or its affiliates), the only DHE nasal product on the market[12]
 - INP104 is a non-oral product that utilizes POD, which delivers DHE to the upper nasal space, increasing systemic absorption of drug compared to lower nasal space delivery[9,12-14]

Objective

- To evaluate within-person consistency of response to INP104 from the STOP 301 study

Methods

Study Design

- STOP 301 was a Phase 3, interventional, open-label, single-group assignment study, assessing the safety, tolerability, and exploratory efficacy of INP104 over 24 and 52 weeks (NCT03557333)
- The study comprised a 4-week screening period, in which patients used their best usual care, a 24-week treatment period for all patients, a treatment extension to 52 weeks for a subset of the patients, and a 2-week post-treatment follow-up period
- Following the screening period, all patients were provided INP104 to nasally self-administer (1.45 mg in a dose of 2 sprays; up to 2 doses/24 hours or 3 doses/7 days) with self-recognized MAs over 24 weeks, with a subset over 52 weeks
- Daily eDiaries were completed to capture headache and migraine details, headache medication usage, and most bothersome symptom severity

Study Patients

- Patients were adult (18–65 years) males or females who had a documented diagnosis of migraine, with a minimum of 2 MAs, with or without aura, each month not qualifying as chronic migraine during the previous 6 months per the *International Classification of Headache Disorders,* version 3 beta (ICHD-3β)
- Patients were in general good health, with no significant medical history or clinical abnormalities at baseline, which included no history of cardiovascular events

Study Outcome Measures

- Post hoc analyses of STOP 301 data were performed on the exploratory efficacy data collected
- Outcomes are reported for the 24-week full safety set (FSS; i.e., all patients who were enrolled and received ≥1 dose of INP104) and are limited to patients with ≥4 INP104-treated MAs in both Weeks 1–12 and Weeks 13–24
- Within-person consistency in 2-hour headache response (2hHR) was defined as the proportion of treated MAs (100%, ≥75%, and ≥67%) having mild or no pain at 2 hours post-INP104 over 24 weeks
 - A patient achieved a 100% consistency threshold if all their MAs had mild pain or no pain at 2 hours, a ≥75% consistency threshold if ≥75% of MAs were mild or pain free, and similarly for the ≥67% threshold
- Results from a patient acceptability questionnaire (PAQ) and the Migraine Disability Assessment (MIDAS) were also examined based on consistency thresholds
- The MIDAS questionnaire consists of 5 scored questions that measure the number of days in the past 3 months on which a patient experienced limitations in daily activities, and was evaluated during the screening period, at enrollment (Day 0), Week 12, and Week 24
 - MIDAS questionnaire grading system: Grade I = minimal or infrequent disability (0–5); Grade II = mild or infrequent disability (6–10); Grade III = moderate disability (11–20); Grade IVa = severe disability (21–40); and Grade IVb = severe disability (41–270)
- A 9-question PAQ asking patients to assess the acceptability, usability, and effectiveness of INP104 was administered at Week 24
 - Results from 3 of these questions will be reported here, and patients responded using a 5-item scale from “strongly agree”, “agree”, “neutral”, “disagree”, or “strongly disagree” (or not applicable)
 - Questions:
 - Compared to previous migraine prescription medications, the investigational product more consistently relieves each one of my migraine headaches
 - With the investigational product I can return to normal activities faster (school/work/leisure activities) compared to my previous prescription migraine medication(s)
 - If the INP104 product were commercially available, I would request a prescription for it from my physician

Results

Consistency of Response

- 188 patients in the 24-week FSS had ≥4 INP104-treated MAs in both Weeks 1–12 and Weeks 13–24, and were included in the consistency analyses
- Over 24 weeks, 63.3% of patients responded in ≥67% of MAs, 59.6% in ≥75% of MAs, and 17.6% in 100% of MAs
- Patients reported the same or improved level of benefit with INP104 during Weeks 13–24 vs Weeks 1–12, indicating within-person consistency
 - During Weeks 1–12 and Weeks 13–24, the proportion of patients that responded in ≥67% of MAs was 64.2% and 63.0%; in ≥75% of MAs, 58.3% and 60.9%; and in 100% of MAs, 25.7% and 30.4%, respectively (**Figure 1**)
 - Of those responding in 100% of MAs during Weeks 1–12, most remained at this threshold during Weeks 13–24 (65.2%)
 - Of those responding in 75 to <100% of MAs during Weeks 1–12, the majority achieved the same or better level of response during Weeks 13–24 (88.5%)

Figure 1. Proportion of Patients Achieving Within-Person Consistency in 2hHR Over 24 Weeks (N=188)

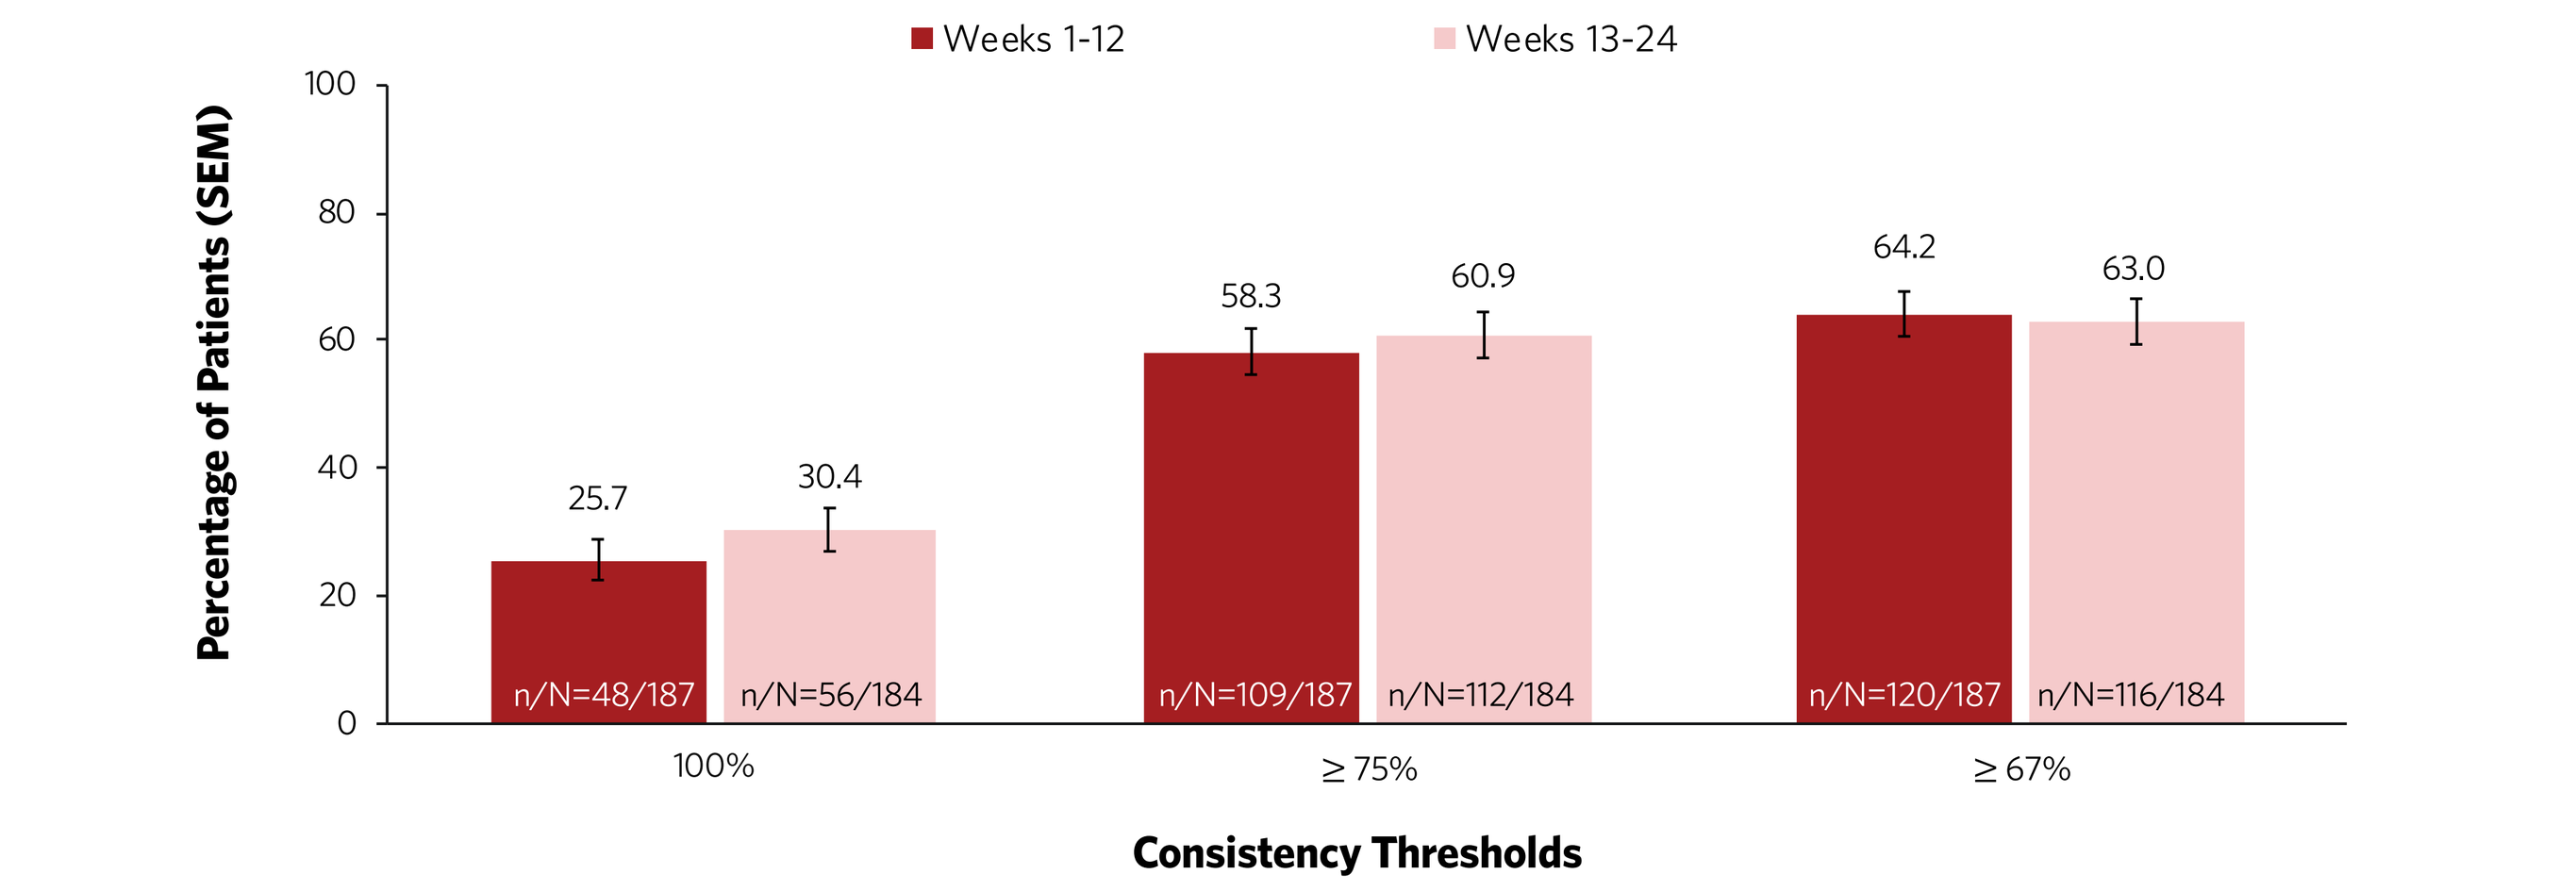
Weeks 1-12
Weeks 13-24
Percentage of Patients (SEM)
100
80
60
40
20
0
25.7
30.4
58.3
60.9
64.2
63.0
n/N=48/187
n/N=56/184
n/N=109/187
n/N=112/184
n/N=120/187
n/N=116/184
100%
≥ 75%
≥ 67%
Consistency Thresholds

Note: Data are self-reported.
2hHR = 2-hour headache response; SEM = standard error of the mean.

PAQ and MIDAS

- Patient acceptability improved and MIDAS scores declined as consistency of response increased
 - Patients with consistency thresholds of 100%, ≥75%, or ≥67% generally strongly agreed/agreed that INP104 consistently relieved their MAs and that they could return to normal activities earlier with INP104 compared to their previous products (**Figure 2**)
 - When patients were asked if they would request a prescription for INP104 from their physician once commercially available, similar rates were reported across the consistency groups (**Figure 2**)
 - At baseline, patients within 100%, ≥75%, and ≥67% consistency thresholds had mean MIDAS scores indicative of severe migraine-specific disability, and clinically relevant improvements in mean MIDAS were observed at Weeks 12 and 24 across all 3 consistency thresholds (**Table 1**)

Figure 2. PAQ Results by 2hHR During Weeks 1–24 (N=188)

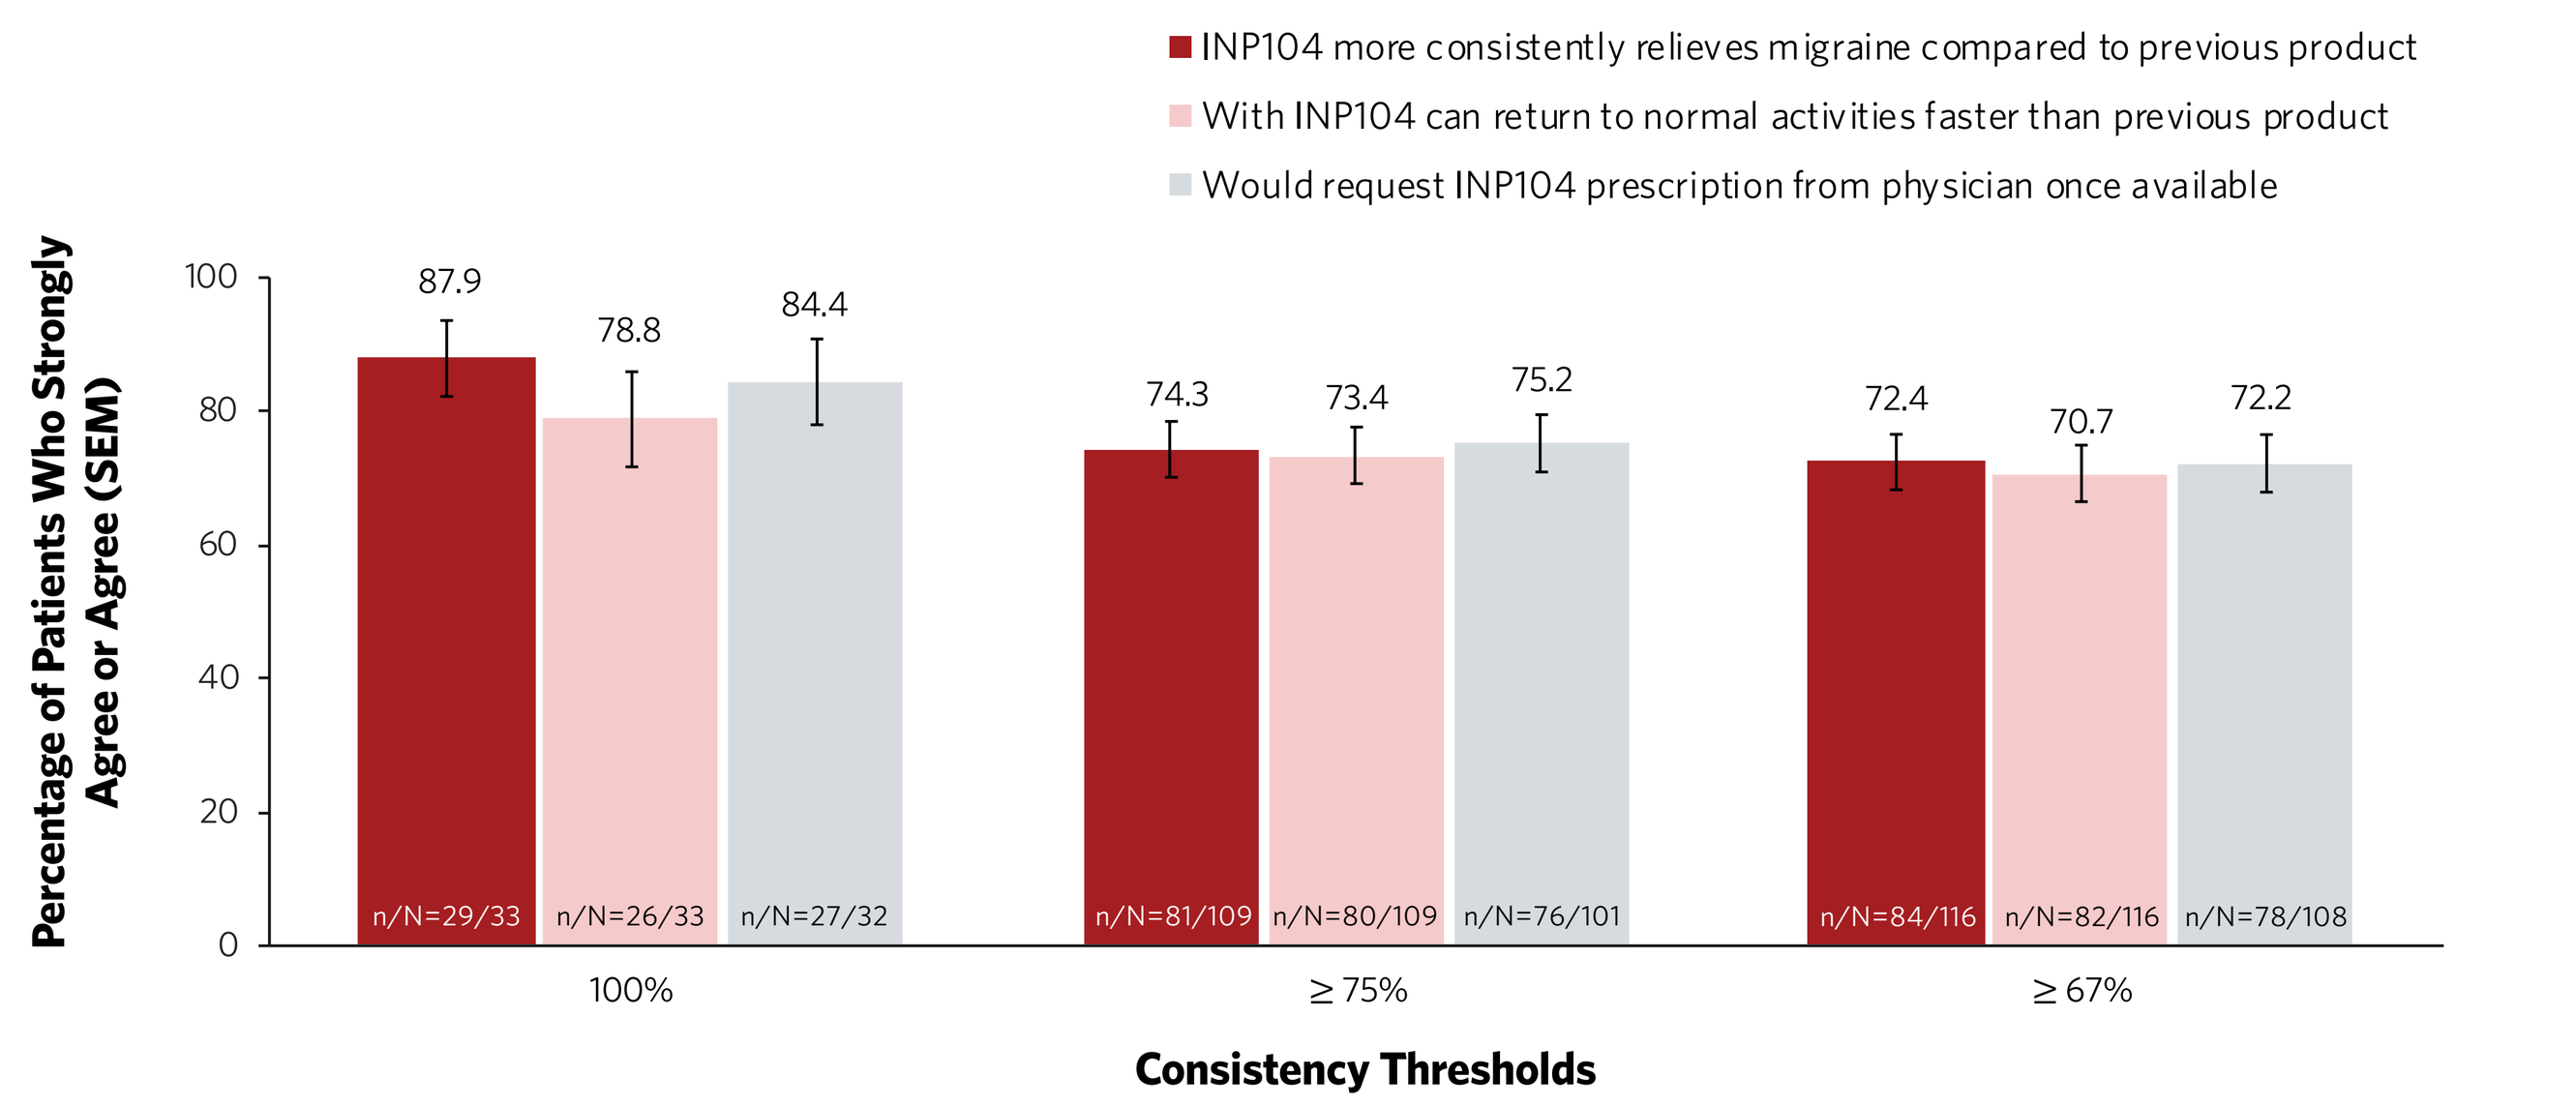
INP104 more consistently relieves migraine compared to previous product
With INP104 can return to normal activities faster than previous product
Would request INP104 prescription from physician once available
Percentage of Patients Who Strongly Agree or Agree (SEM)
100
80
60
40
20
0
87.9
78.8
84.4
74.3
73.4
75.2
72.4
70.7
72.2
n/N=29/33
n/N=26/33
n/N=27/32
n/N=81/109
n/N=80/109
n/N=76/101
n/N=84/116
n/N=82/116
n/N=78/108
100%
≥ 75%
≥ 67%
Consistency Thresholds

2hHR = 2-hour headache response; PAQ = patient acceptability questionnaire; SEM = standard error of the mean.

Table 1. MIDAS Scores by 2hHR During Weeks 1–24 (N=188)

Period		Consistency Thresholds 100%	≥75%	≥67%
Baseline	n	33	112	119
	Mean (SD)	23.9 (25.35)	26.0 (23.31)	25.3 (22.86)
Week 12	n	32	111	118
	Mean (SD)	20.9 (25.00)	19.1 (18.83)	19.0 (18.39)
	Change from baseline, mean (SD)	-3.2 (21.38)	-7.0 (19.52)	-6.3 (19.25)
Week 24	n	33	112	119
	Mean (SD)	17.2 (20.44)	17.7 (18.45)	17.7 (18.11)
	Change from baseline, mean (SD)	-6.7 (15.45)	-8.3 (18.00)	-7.6 (17.72)

2hHR = 2-hour headache response; MIDAS = Migraine Disability Assessment; SD = standard deviation.

Conclusions

- Results demonstrate that INP104 provided within-person consistency for a 2hHR across multiple MAs over 24 weeks, which was also associated with improvements in patient acceptability (PAQ) and reductions in disability
- Data presented here may reflect a real-world setting by demonstrating that if a patient chooses to adhere to INP104 over 24 weeks, they can likely expect consistency of response
 - Reasons why patients chose not to continue INP104 treatment in the STOP 301 study included adverse events (6.8%) or lack of efficacy (5.9%)[15] or that treatment with INP104 was associated with decreased migraine frequency as the study progressed (results to be reported in future disclosure)
- The consistency of INP104 may be attributed to several factors, including the more consistent systemic availability due to absorption of DHE by POD delivery to the upper nasal space; the familiar pharmacology of IV DHE, which has a long-standing reputation for being efficacious; and the non-oral route of delivery that bypasses the gut[9-14]
- Findings also align with previously published STOP 301 exploratory efficacy data indicating that the single-attack efficacy of INP104 was sustained with intermittent usage over multiple MAs through 24 weeks[15]

References

1. Lipton RB, et al. *Cephalalgia.* 2009;29:826-836.
2. McGinley JS, et al. *Headache.* 2019;59:1002-1013.
3. Leroux E, et al. *Adv Ther.* 2020;37:4765-4796.
4. Lombard L, et al. *J Headache Pain.* 2020;21:41.
5. Kori S, et al. *J Headache Pain.* 2013;14(Suppl 1):P190.
6. Tokola RA, Neuvonen PJ. *Br J Clin Pharmacol.* 1984;18:867-871.
7. Aurora SK, et al. *Headache.* 2006;46:57-63.
8. Camara-Lemarroy CR, et al. *World J Gastroenterol.* 2016;22:8149-8160.
9. Rapoport AM, et al. *CNS Drugs.* 2010;24:929-940.
10. Silberstein SD, et al. *Headache.* 2020;60:40-57.
11. Baron EP, Tepper SJ. *Future Neurol.* 2011;6:327-333.
12. Shrewsbury SB, et al. *Headache.* 2019;59:394-409.
13. Hoekman J, et al. *US Neurol.* 2020;16:25-31.
14. Homayun B, et al. *Pharmaceutics.* 2019;11:129.
15. Aurora S, et al. *Cephalalgia.* 2020;40(Suppl 1):12-13. MTV20-OR-015.

Disclosures and Acknowledgments

Richard B. Lipton receives research support from the NIH and the FDA as well as the National Headache Foundation and the Marx Foundation. He also receives research support from Allergan/AbbVie, Amgen, Eli Lilly, and electroCore. He receives personal fees as a consultant or advisor from Allergan/AbbVie, Amgen, Biohaven Pharmaceutical Holding Co. Ltd., Dr. Reddy's, GlaxoSmithKline, Grifols, Impel NeuroPharma, Lundbeck, Merck, Novartis, and Teva Pharmaceuticals. He holds stock or options in Biohaven Holdings and Ctrl M Health. In addition, he receives royalties for *Wolff's Headache and Other Head Pain,* 7th and 8th editions. Barbara L. Nye has participated in Advisory Boards for Alder Biopharmaceuticals, Allergan, Biohaven Pharmaceuticals, electroCore, Pernix, Impel NeuroPharma, and Xoc Pharmaceuticals, Inc. She has been a clinical trial site principal investigator for Allegran, Amgen, ATI, electroCore, Novartis, Satsuma Pharmaceuticals, and Teva Pharmaceuticals. Sheena K. Aurora and Stephen B. Shrewsbury are full-time employees of Impel NeuroPharma and are stockholders in Impel NeuroPharma. Stephen B. Shrewsbury is an officer of Impel NeuroPharma. Joe Hirman is a paid consultant to Impel NeuroPharma. This research was sponsored by Impel NeuroPharma. Editorial support was provided by IMPRINT Science and funded by Impel NeuroPharma. IMPEL, POD, and the IMPEL Logo are registered trademarks of Impel NeuroPharma.

Impel NEUROPHARMA®

Presented at the Virtual AHS Annual Scientific Meeting, June 3–6, 2021