Acute Treatment of Migraine With INP104: Exploratory Efficacy From the Phase 3 STOP 301 Study

Timothy R. Smith, MD[1]; Sheena Aurora, MD[2]*; Jasna Hocevar-Trnka, MD, MPH[2]; Stephen Shrewsbury, MB ChB[2]

[1]StudyMetrix Research, St. Peters, MO, USA; [2]Impel NeuroPharma, Seattle, WA, USA

*Presenting author

Introduction

- Patients continue to report dissatisfaction with acute therapies for migraine[1,2]
- Gastrointestinal (GI) symptoms and comorbidities frequently accompany migraine and may affect the absorption of oral medications and/or lead to patient reluctance to use oral therapies[3-7]
- INP104, an investigational drug-device product that targets dihydroergotamine mesylate (DHE) to the upper nasal cavity using Precision Olfactory Delivery (POD®) technology, is a nasal product that bypasses the GI tract and increases systemic absorption of drug compared to traditional nasal delivery[3,8,9]
- Results from a Phase 1 study (STOP 101) showed that INP104 approached intravenous (IV) DHE-like blood levels from 20 minutes to 48 hours with a lower C_{max} and incidence of adverse events (AEs) than IV DHE[8]
- While the primary focus of the pivotal STOP 301 study was to assess upper nasal space safety and tolerability, which was previously presented,[10] it also included exploratory efficacy of INP104 for the acute treatment of migraine

Objective

- To report exploratory, self-reported efficacy data for INP104 from the STOP 301 study

Methods

Study Design

- This was a Phase 3, interventional, open-label, single-group assignment study, assessing the safety, tolerability, and exploratory efficacy of INP104 (NCT03557333)
- The study comprised a 4-week screening period, where patients used their best usual care, a 24-week treatment period for all patients, a treatment extension to 52 weeks for a subset of the patients, and a 2-week post-treatment follow-up period (**Figure 1**)
- Following screening, all patients were provided with up to 3 doses/week of INP104 to nasally self-administer (1.45 mg in a dose of 2 sprays) with self-recognized migraine attacks (MAs) over 24 weeks, with a subset over 52 weeks
- Daily eDiaries were completed to capture headache and migraine details, headache medication usage, and most bothersome symptom (MBS) severity from screening through 24 weeks and, if applicable, 52 weeks

Figure 1. Study Design

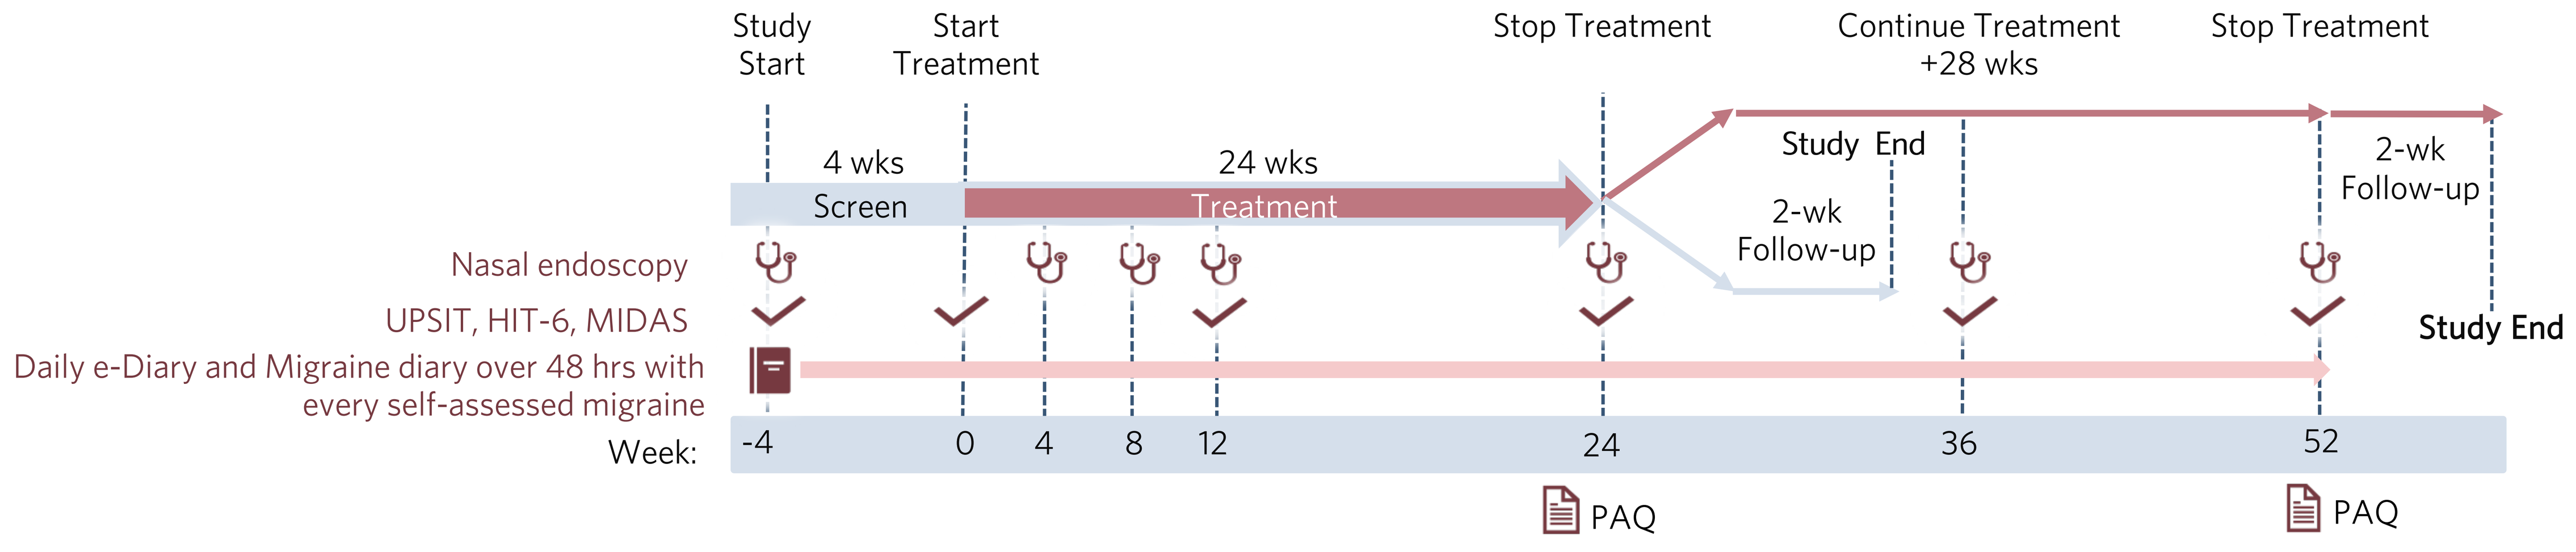
Study Start
Start Treatment
Stop Treatment
Continue Treatment +28 wks
Stop Treatment
4 wks
Screen
24 wks
Treatment
Study End
2-wk Follow-up
2-wk Follow-up
Study End
Nasal endoscopy
UPSIT, HIT-6, MIDAS
Daily e-Diary and Migraine diary over 48 hrs with every self-assessed migraine
Week:
-4
0
4
8
12
24
36
52
PAQ
PAQ

HIT-6 = Headache Impact Test-6; MIDAS = Migraine Disability Assessment; PAQ = patient acceptability questionnaire; UPSIT = University of Pennsylvania Smell Identification Test; wk = week.

Study Patients and Assessments

- Patients were adult (18–65 years) males or females with a documented diagnosis of migraine, with a minimum of 2 MAs, with or without aura, each month not qualifying as chronic migraine during the previous 6 months per the *International Classification of Headache Disorders,* version 3 beta
- Patients were in general good health, with no significant medical history or clinical abnormalities at baseline, which included no history of cardiovascular events
- Self-reported exploratory endpoints included pain and MBS freedom at 2 hours, sustained pain freedom at 24 and 48 hours, use of rescue medication, and within-person consistency of response across multiple MAs post-INP104 over 24 and 52 weeks
- Within-person consistency in 2-hour headache response was defined as the proportion of treated MAs (100%, ≥75%, and ≥67%) having mild or no pain at 2 hours post-INP104
- Statistical comparisons to best usual care during baseline were not performed because patients were permitted to administer acute therapies (sometimes more than one) of their choosing to treat their MAs during baseline

Results

Patient Disposition and Baseline Characteristics

- 360 patients were screened and enrolled into the 24-week treatment period
 - 354 patients received ≥1 dose of INP104 (24-week full safety set [FSS])
 - 73 patients continued into the 28-week extension period (52-week FSS)
- 262 and 66 patients completed the 24- and 52-week treatment periods, respectively

Pain and MBS Freedom

- During Weeks 1–12, patients treated 2,559 MAs with INP104, and 39% and 55% of MAs were pain- and MBS-free at 2 hours, respectively
- During Weeks 13–24, patients treated 1,736 MAs with INP104, and 35% and 51% of MAs were pain- and MBS-free at 2 hours, respectively
- Results for the 52-week period were similar (**Figure 2**)
- Compared to baseline, more MAs were pain- and MBS-free 2 hours post-INP104 at each 4-week interval over 24 and 52 weeks
 - Mean number of MAs self-reported as pain- and MBS-free 2-hours post-INP104 ranged from 35.1–38.7% and 49.2–57.9% compared to 30.6% and 47.9% at baseline, respectively (24-week FSS)
 - Mean number of MAs self-reported as pain- and MBS-free 2-hours post-INP104 ranged from 31.4–39.0% and 39.8–55.7% compared to 23.5% and 40.8% at baseline, respectively (52-week FSS)

Sustained Pain Freedom

- 37% of MAs were pain-free at 2 hours post-INP104, and 35% and 32% had sustained pain freedom at 24 and 48 hours, respectively, over 24 weeks (**Figure 3**)

Figure 2. Pain and MBS Freedom at 2 Hours Post-INP104 by 12-week Intervals (52-week FSS)

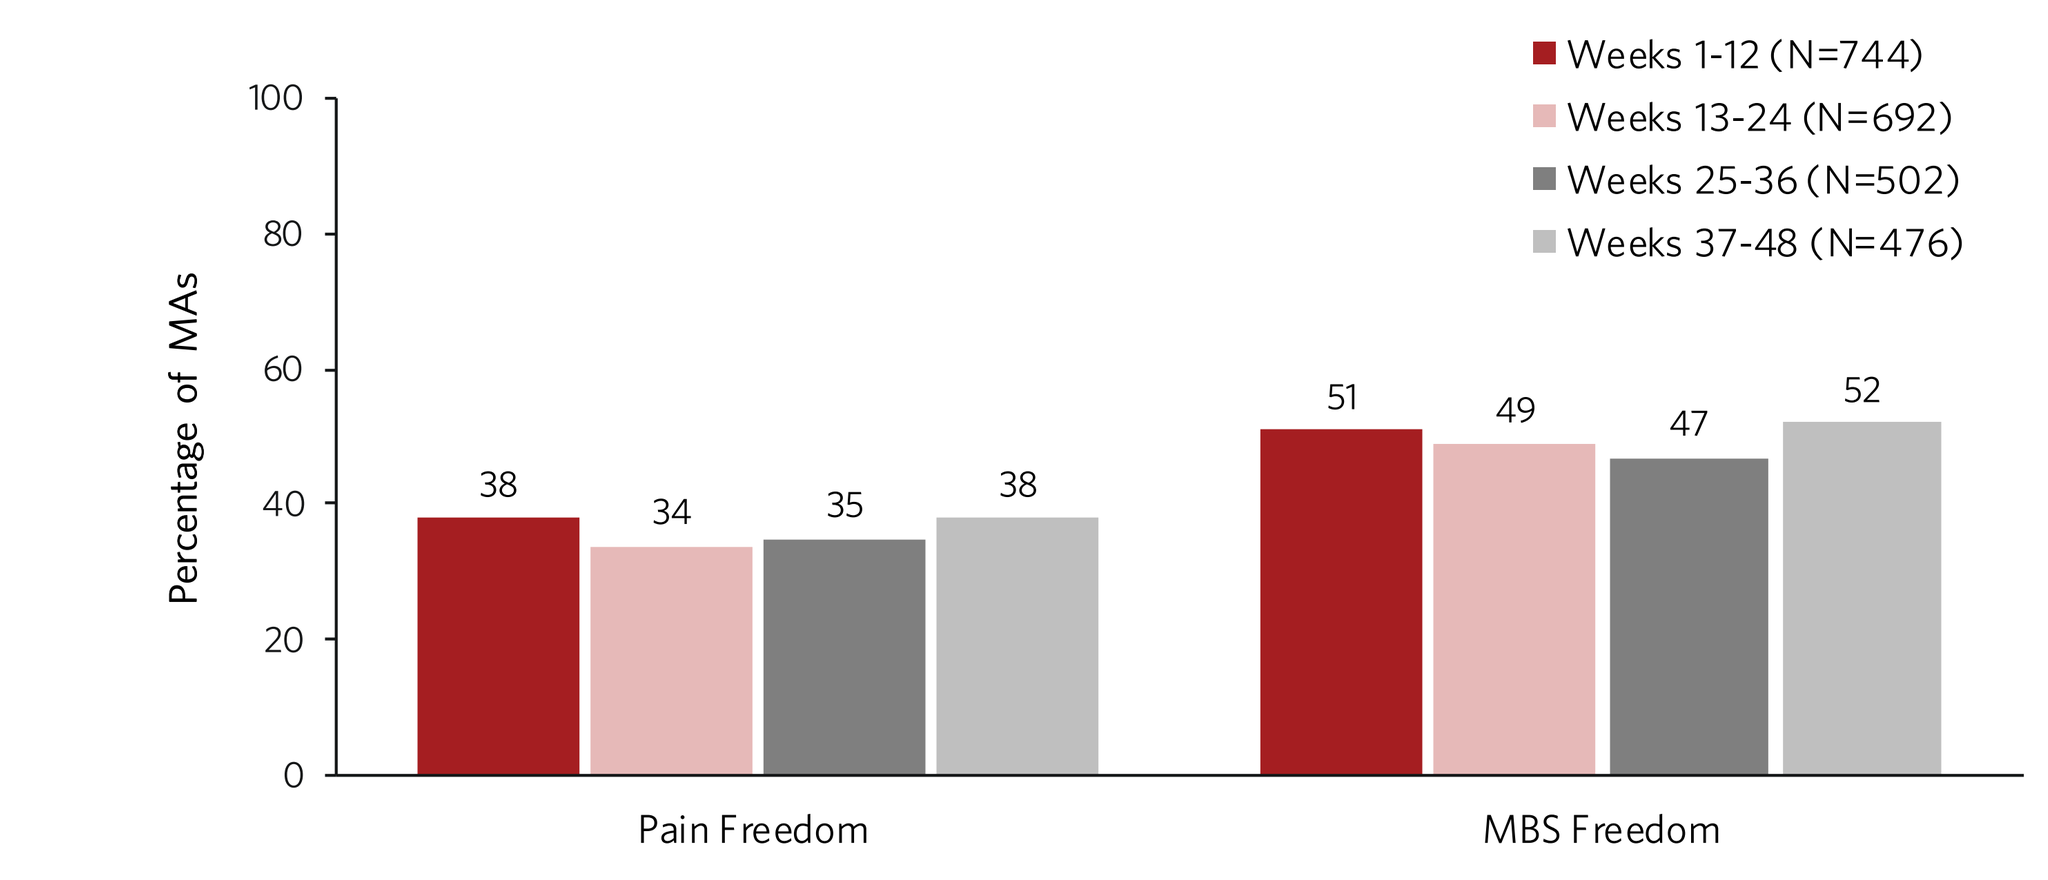
Weeks 1-12 (N=744)
Weeks 13-24 (N=692)
Weeks 25-36 (N=502)
Weeks 37-48 (N=476)
Percentage of MAs
100
80
60
40
20
0
38
34
35
38
51
49
47
52
Pain Freedom
MBS Freedom

Note: Data are self-reported. N is the number of INP104-treated MAs with a non-missing 2-hour assessment.

FSS = full safety set; MA = migraine attack; MBS = most bothersome symptom.

Figure 3. Pain- and Sustained-Pain Freedom Rates (24-week FSS)

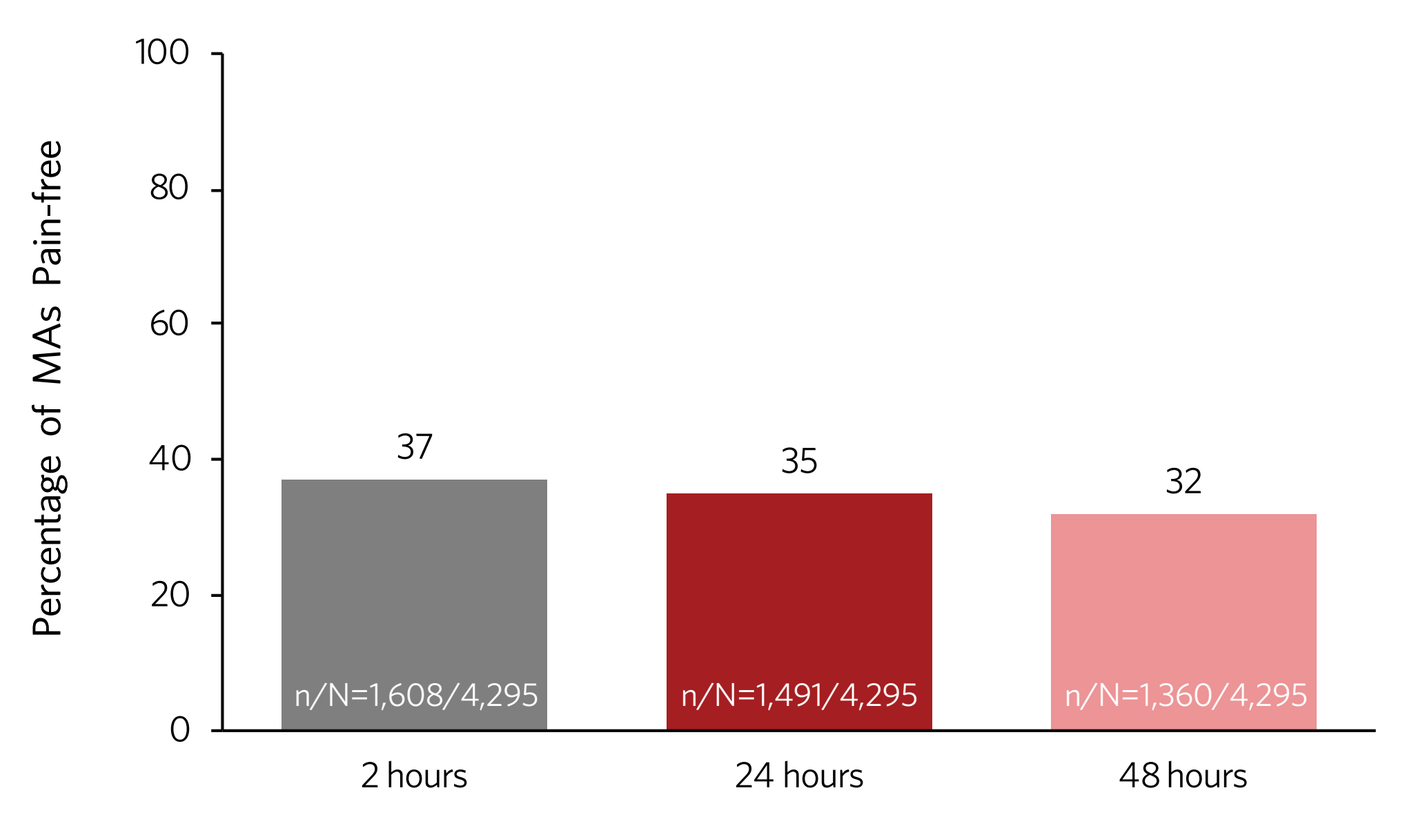
Percentage of MAs Pain-free
100
80
60
40
20
0
37
35
32
n/N=1,608/4,295
n/N=1,491/4,295
n/N=1,360/4,295
2 hours
24 hours
48 hours

Note: Data are self-reported. Counts are MAs with a non-missing assessment.

FSS = full safety set; MA = migraine attack.

Rescue Medication

- 15% of MAs required rescue medication over 24 weeks (**Figure 4**)
- 90.9% used non-INP104 medications (a second INP104 dose was allowed within 24 hours)

Consistency of Response

- Over 24 weeks, 25.0%, 57.0%, and 59.9% of patients were 100%, ≥75%, and ≥67% responders, respectively
- After 24 weeks, 22.2%, 58.3%, and 65.3% of patients were 100%, ≥75%, and ≥67% responders, respectively (**Figure 5**)

Figure 4. Rescue Medication Use (N=4,257 MAs; 24-week FSS)

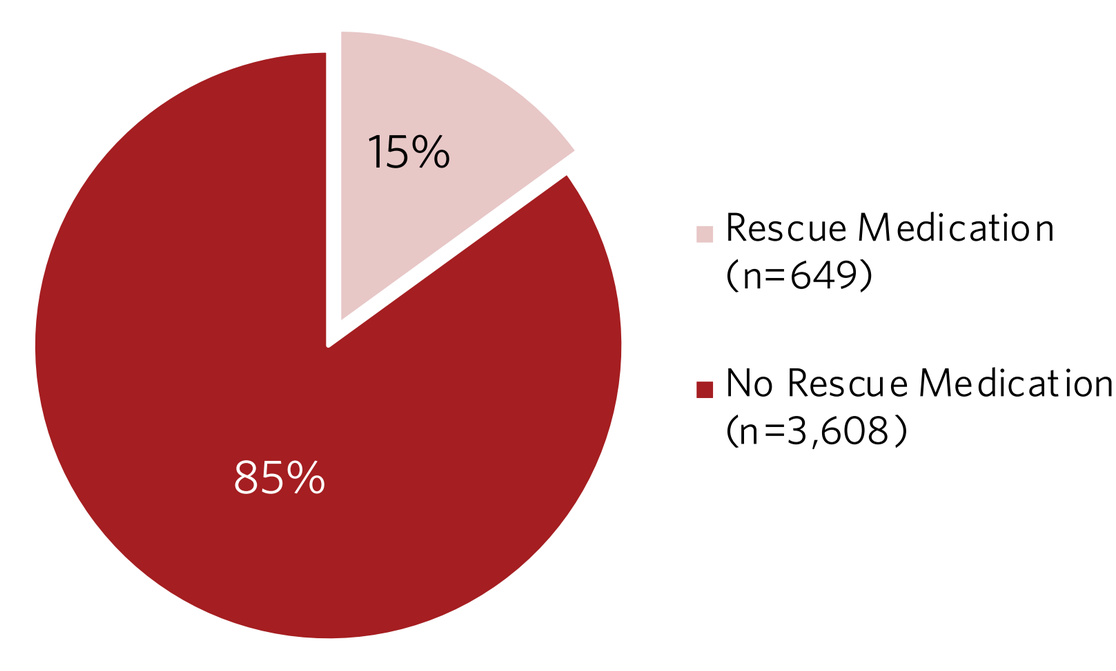
15%
85%
Rescue Medication (n=649)
No Rescue Medication (n=3,608)

Note: Data are self-reported. Excludes MAs that started at baseline and ended in Weeks 1–24 with acute medication use in both periods.

FSS = full safety set; MA = migraine attack.

Figure 5. Within-Person Consistency for Mild or No Pain at 2 Hours Post-INP104 (FSS)

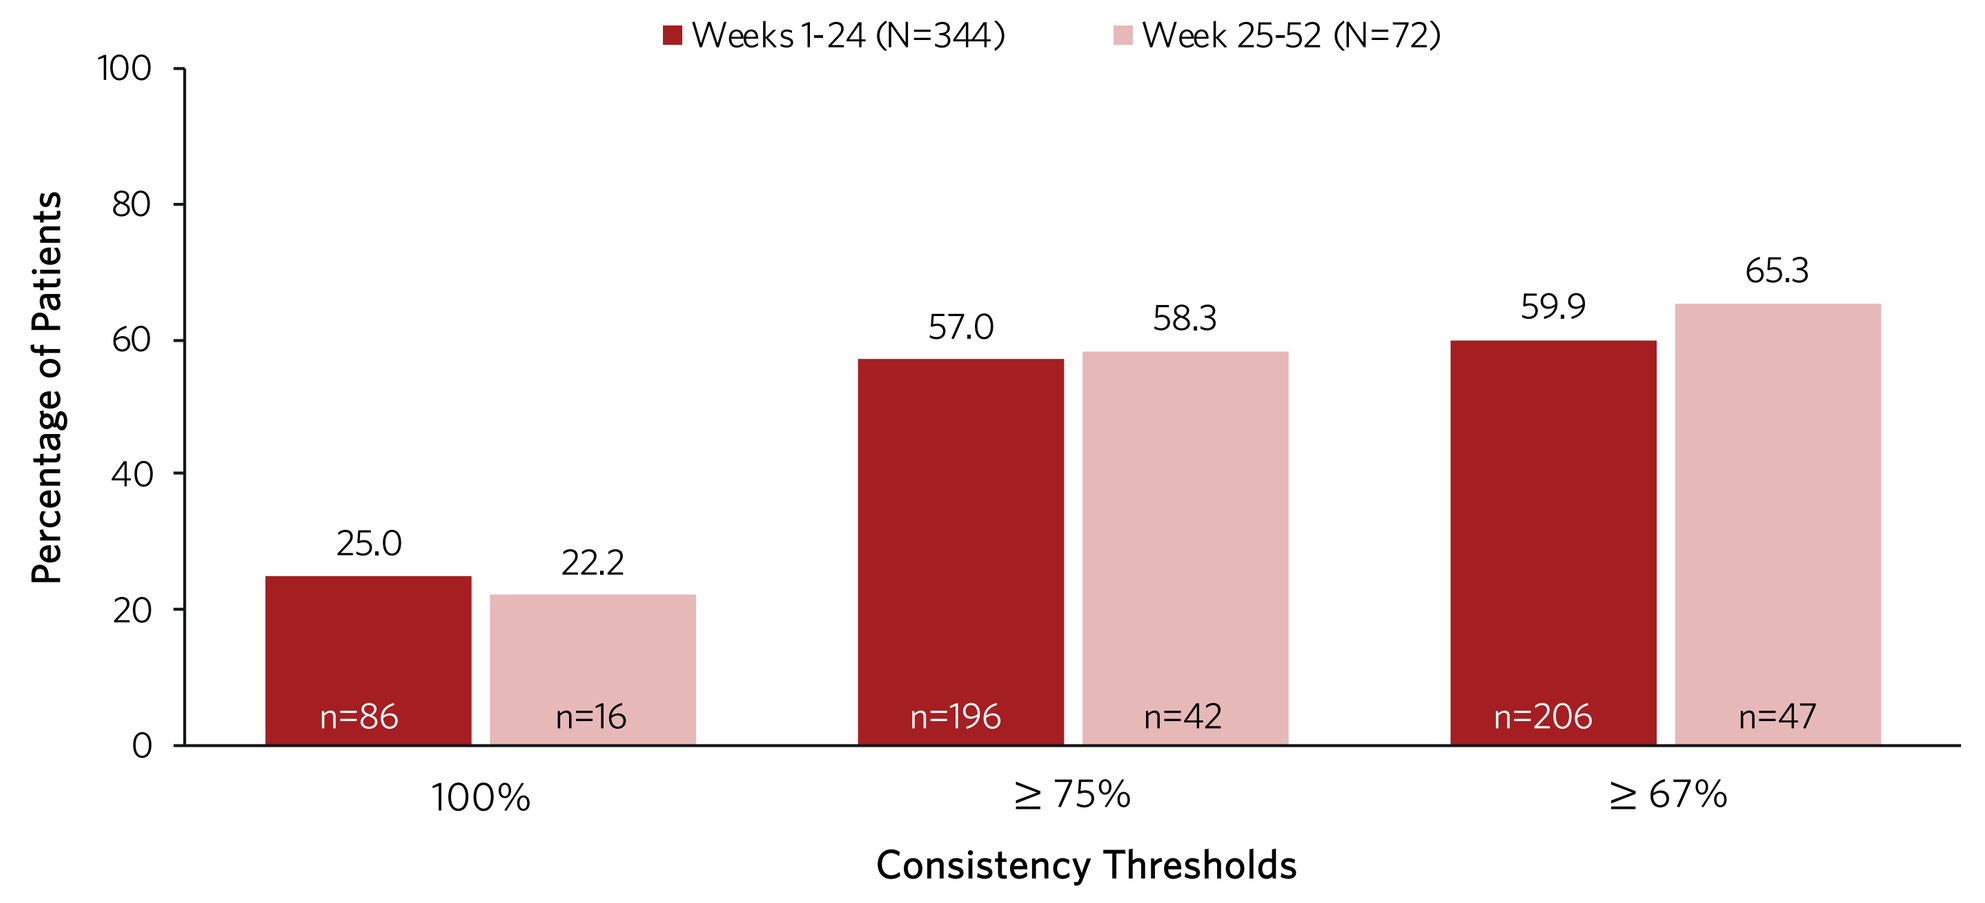
Weeks 1-24 (N=344)
Week 25-52 (N=72)
Percentage of Patients
100
80
60
40
20
0
25.0
22.2
57.0
58.3
59.9
65.3
n=86
n=16
n=196
n=42
n=206
n=47
100%
≥ 75%
≥ 67%
Consistency Thresholds

Note: Data are self-reported.

FSS = full safety set.

Conclusion

- STOP 301 was an open-label study of safety, tolerability, and exploratory efficacy of long-term intermittent usage of nasal DHE (INP104) self-administered over 24 and 52 weeks
- The use of INP104 was associated with improvements in several migraine measures of exploratory efficacy
- In the absence of a direct comparison, results suggest that INP104 was associated with high rates of symptom freedom and may be a promising new acute treatment for migraine

References

1. Lipton RB, et al. *Headache.* 2019;59:1310-1323.
2. Hutchinson S, et al. *Mayo Clin Proc.* 2020;95:709-718.
3. Rapoport AM, et al. *CNS Drugs.* 2010;24:929-940.
4. Aurora SK, et al. *Headache.* 2006;46:57-63.
5. Hindiyeh N, et al. *Curr Pain Headache Rep.* 2015;19:33.
6. Aurora SK, et al. *Cephalalgia.* 2013;33:408-415.
7. Camara-Lemarroy CR, et al. *World J Gastroenterol.* 2016;22:8149-8160.
8. Shrewsbury SB, et al. *Headache.* 2019;59:394-409.
9. Hoekman J, et al. *US Neurol.* 2020;16:25-31.
10. Aurora SK, et al. *Cephalalgia.* 2020;40(Suppl 1):3-17. MTV20-OR-015.

Disclosures and Acknowledgments

Timothy R. Smith has supported clinical trial research for Lundbeck, Amgen, Allergan/AbbVie Inc., Biohaven Pharmaceuticals, Boehringer Ingelheim, Charleston Laboratories, Inc., electroCore, Inc., Impel NeuroPharma, Eli Lilly and Company, Pfizer Inc., Novartis, Novo Nordisk, Satsuma Pharmaceuticals, Inc., Teva Pharmaceutical Industries Ltd., Theranica Bio-Electronics Ltd., and Vorso Corp. He has participated in speaker bureaus for Amgen, Allergan/AbbVie Inc., Biohaven Pharmaceuticals, and Eli Lilly and Company, and is an advisor/consultant for Lundbeck, Amgen, Allergan/AbbVie Inc., Biohaven Pharmaceuticals, Impel NeuroPharma, Eli Lilly and Company, Theranica Bio-Electronics Ltd., and Vorso Corp. He is also a stockholder in the UnitedHealth Group and an officer and Board of Directors member (unpaid volunteer work for a nonprofit organization) for the National Headache Foundation. Sheena K. Aurora, Stephen B. Shrewsbury, and Jasna Hocevar-Trnka are full-time employees and stockholders of Impel NeuroPharma. This research was sponsored by Impel NeuroPharma. Editorial support was provided by IMPRINT Science and funded by Impel NeuroPharma. IMPEL, POD, and the IMPEL Logo are registered trademarks of Impel NeuroPharma.

Presented at the Virtual AHS Annual Scientific Meeting, June 3–6, 2021