Nasal Safety of Chronic Intermittent Use of INP104: Results From the Phase 3 Open-label STOP 301 Study

Greg Davis, MD, MPH[1]; Adil Fatakia, MD[2]; Seth Pransky, MD[3]; Alison E. Heald, MD[4]; Sheena K. Aurora, MD[4]; Jasna Hocevar-Trnka, MD, MPH[4]; Stephen B. Shrewsbury, MB ChB[4]*

[1]Proliance Surgeons, Seattle, WA, USA; Ear Nose Throat & Allergy Associates, Puyallup, WA, USA; [2]New Orleans Sinus Center, New Orleans, LA, USA; [3]Pediatric Otolaryngology, Head and Neck Surgery, San Diego, CA, USA; [4]Impel NeuroPharma, Seattle, WA, USA

*Presenting author

Introduction

- INP104 is an investigational, drug-device product that delivers dihydroergotamine mesylate to the upper nasal space by Precision Olfactory Delivery (POD®) technology for the acute treatment of migraine[1,2]
- The STOP 301 study was designed to evaluate the safety, tolerability, and exploratory efficacy of INP104 over 24 or 52 weeks, and top-line data were previously presented[3]
- Primary endpoints were the number of patients with serious and non-serious treatment-emergent adverse events (TEAEs), change in nasal mucosal integrity observed on endoscopy, and change in olfactory function
- Since a standardized objective safety assessment instrument for the upper nasal space was not previously available, Impel NeuroPharma worked closely with experts in the field to design appropriate scales to assess changes to the upper nasal space in the STOP 301 study
- The following tools were employed:
 - University of Pennsylvania Smell Identification Test (UPSIT), a validated tool for olfactory function assessment[4]
 - Quantitative Scoring Scale for Evaluation of the Nasal Mucosa (QSS-NM) to categorize changes to the upper nasal mucosa, a novel scale adapted from the modified Lund-Kennedy score (MLKS) that is familiar to otolaryngologists[5]

Objective

- To assess the safety and tolerability of long-term intermittent use of INP104 on the nasal mucosa and olfaction for 24 and 52 weeks in the STOP 301 study

Methods

Study Design

- STOP 301 was a Phase 3, interventional, open-label, single-group assignment study (NCT03557333)
- 4-week screening period, in which patients used their best usual care to treat migraine attacks (MAs), a 24-week treatment period for all patients, a treatment extension to 52 weeks for a subset of the patients, and a 2-week post-treatment follow-up period
- Following the screening period, all patients were provided with up to 3 doses/week of INP104 to nasally self-administer (1.45 mg in a dose of 2 sprays) with self-recognized MAs

Study Patients

- Adults with a diagnosis of migraine, with a minimum of 2 MAs, with or without aura, each month without qualifying as chronic migraine (≥15 MA days/month) during the previous 6 months

Study Outcome Measures

- A Nasal Safety Review Committee (NSRC), consisting of 3 independent otolaryngologists, participated in the development and evaluation of nasal safety monitoring tools
- UPSIT
 - 40-item scratch-and-sniff test performed approximately every 12 weeks
 - A significant change in the UPSIT score was defined as a ≥5-point reduction based on the best current evidence
 - Asymptomatic ≥5-point UPSIT score reductions were recorded as the TEAE "olfactory test abnormal"
- Nasal endoscopic examinations (**Figure 1**)
 - Performed by an otolaryngologist; scored using QSS-NM to measure mucosal irritation, epistaxis, mucosal edema, nasal discharge, and mucosal crusting
 - Performed before the 2nd visit (end of baseline) and at 4, 8, 12, and 24 weeks, and 36 and 52 weeks for extension
 - A total score >7 (left + right upper nasal scores), or an individual score ≥2 on any item at any time point triggered a repeat examination 2 weeks later
- Nasal AEs, defined as AEs associated with the nose in any way, were identified using a custom Medical Dictionary for Regulatory Activities (MedDRA) query list

Figure 1. Nasal Endoscopy

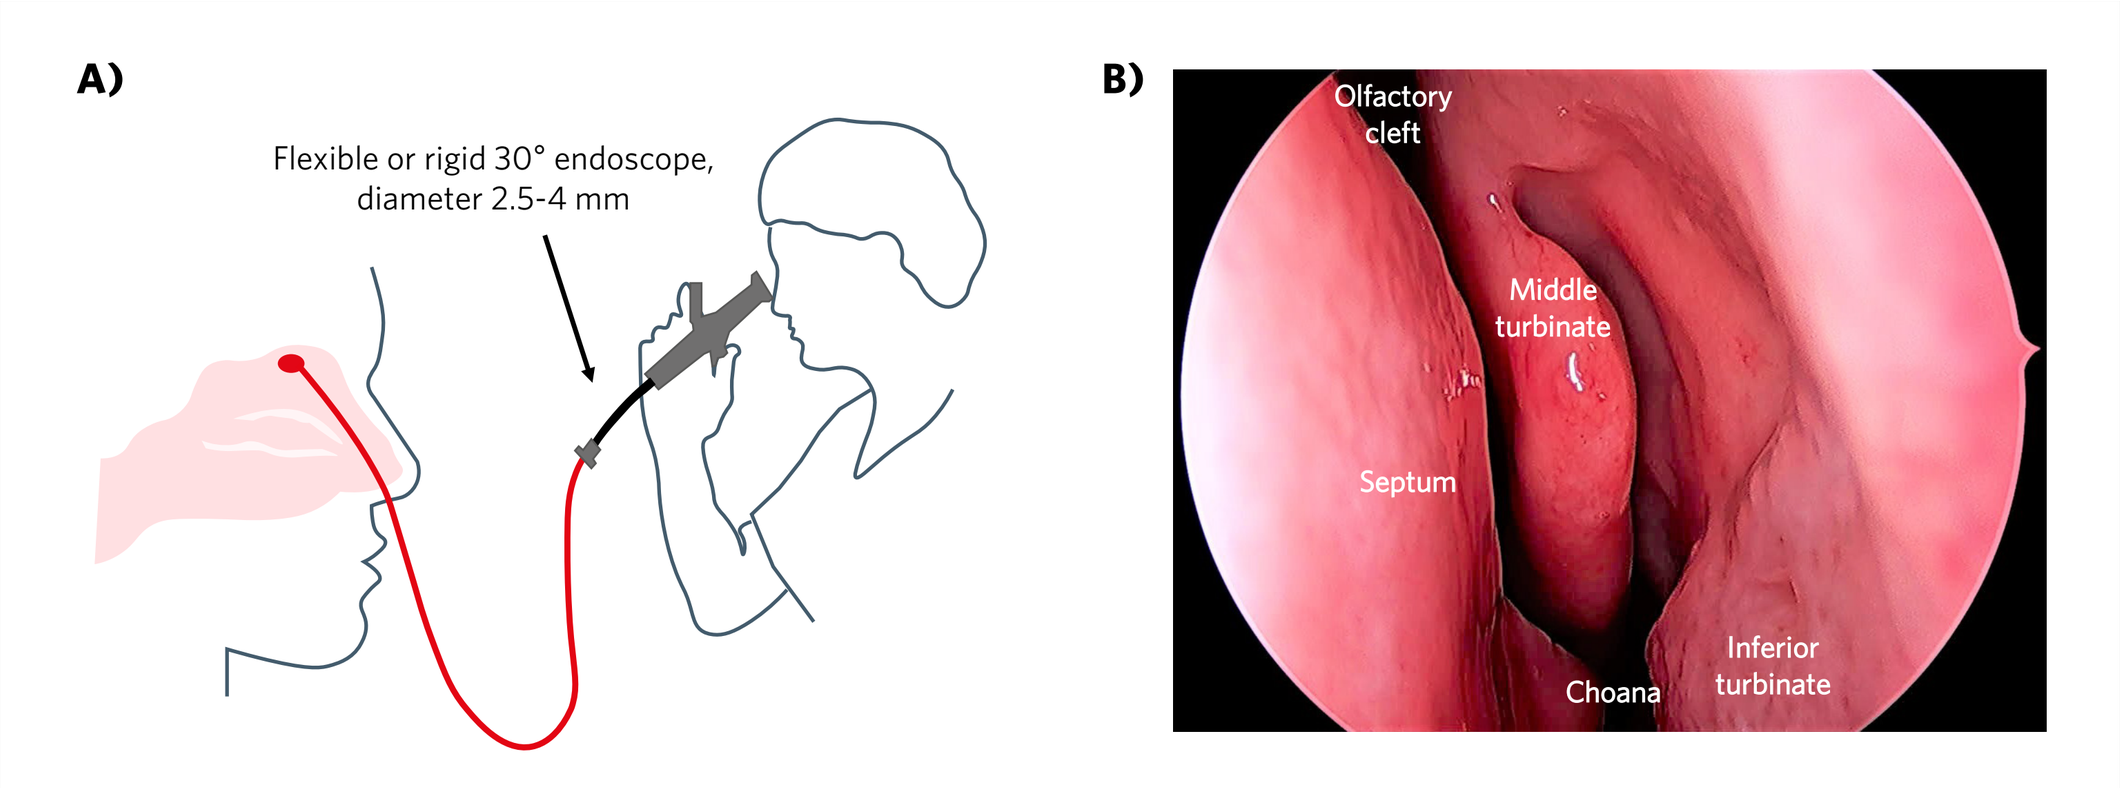
A)
Flexible or rigid 30° endoscope, diameter 2.5-4 mm
B)
Olfactory cleft
Middle turbinate
Septum
Choana
Inferior turbinate

A) Nasal endoscopy process; B) Diagram of normal nasal endoscopy (courtesy of Dr. Davis).

Results

Patient Demographics and Disposition

- 360 patients enrolled, 354 treated ≥1 MA (24-week full safety set [FSS]), and 262 completed 24 weeks of treatment
- 73 patients entered the 28-week extension (52-week FSS), with 66 completing 52 weeks of treatment
- 6,332 INP104 doses were self-administered over 52 weeks

Nasal TEAEs

- 45.8% (162/354) and 58.9% (43/73) of patients reported a nasal TEAE in the 24- and 52-week FSS, respectively, of which none was serious
- Most common (≥10%) were nasal congestion (16.7%; n=59) and upper respiratory tract infection (URTI; 10.7%; n=38) in the 24-week FSS
- Most were mild or moderate, with a single severe case of nasal congestion
- Led to INP104 discontinuation in 4% (14/354) and 1.4% (1/73) of patients during the 24- and 52-week treatment periods, respectively
 - Most frequent were nasal congestion (1.4%; 5/354) and nasal discomfort (1.1%; 4/354) in the 24-week FSS
- No increase in TEAE frequency was observed with more frequent or longer duration of INP104 dosing

Olfactory Function Changes (UPSIT)

- Minimal mean decreases (<0.5) and mean increases/decreases (≤1.0) from baseline were observed in the UPSIT score for the 24- and 52-week FSS at all time points, respectively (**Table 1**)

Table 1. Change From Baseline in UPSIT Score by Visit

Period		24-week FSS (N=354)	52-week FSS (N=73)
Baseline[a]	n	354	73
	Mean (SD)	35.25 (2.998)	35.08 (2.915)
Week 12[b]	n	280	72
	Mean (SD)	34.82 (3.854)	34.88 (3.642)
	Change from baseline[c], mean (SD)	-0.48 (2.690)	-0.18 (2.573)
Week 24[b]	n	206	55
	Mean (SD)	35.03 (3.182)	34.95 (2.599)
	Change from baseline[c], mean (SD)	-0.22 (2.270)	0.03 (2.373)
Week 36[b]	n	—	70
	Mean (SD)		34.89 (3.618)
	Change from baseline[c], mean (SD)		-0.13 (2.930)
Week 52[b]	n	—	65
	Mean (SD)		34.12 (4.106)
	Change from baseline[c], mean (SD)		-0.80 (2.710)

[a]Baseline was defined as the average of UPSIT scores at screening and baseline; hence, if one of these results was missing, baseline was the result from the other time point; [b]Post-baseline only included data from patients who started the first INP104 dose on/before the visit evaluated; [c]For change from baseline summaries, only patients with a value at both baseline and a post-baseline visit were included.

FSS = full safety set; SD = standard deviation; UPSIT = University of Pennsylvania Smell Identification Test.

Nasal Endoscopy Findings

- Mean increases from baseline in QSS-NM total score were minimal (≤0.2) at each post-baseline visit up to 52 weeks (**Table 2**)
 - 4 patients in the 24-week FSS had a total QSS-NM score change of >7 points, while 12 had a change of ≥2 on any 1 of the 5 individual QSS-NM scores
 - Individual scores were 0 for >94% and >92% of patients at baseline and post-baseline visits, respectively, for the 24-week FSS
- Over 90% of patients had normal upper nasal endoscopies over 24 weeks
 - Abnormal, clinically significant changes were noted in 4 patients at Week 4 and 3 patients at Weeks 8 and 12 (**Table 3**)

Nasal TEAEs Associated With Nasal Endoscopy and UPSIT Findings

- A summary of nasal TEAEs, nasal endoscopy findings, and UPSIT score decreases meeting protocol-specified shift criteria is depicted in **Figure 2**
- 23 patients experienced 26 TEAEs associated with abnormal clinically significant upper endoscopy or QSS-NM shift criteria
 - 1 additional patient met the nasal endoscopy shift criteria due to an ongoing pre-existing medical condition of seasonal allergies but with no associated nasal TEAE
 - 22 of 26 cases associated with TEAEs were considered unlikely to be related to INP104
- 25 patients experienced 28 TEAEs associated with an UPSIT score decrease of ≥5 points
 - 17 TEAEs were asymptomatic "olfactory test abnormal," 9 were associated with URTI or sinus symptoms, and 2 were mild hyposmia without other URT symptoms
 - 16 patients had resolution of the UPSIT score decrease from baseline at the first repeat test performed, with 5 events ongoing as of last patient contact (4 events had not resolved at follow-up and 1 was lost to follow-up)
 - The remainder of events resolved, recurred, and then resolved again (or in 1 case they were withdrawn)

Table 2. Change From Baseline in Upper Nasal Endoscopy by QSS-NM Total Score by Visit

Period		24-week FSS (N=354)	52-week FSS (N=73)
Baseline[a]	n	354	73
	Mean (SD)	0.1 (0.39)	0.2 (0.53)
Week 4[b]	n	308	71
	Mean (SD)	0.3 (1.31)	0.2 (0.62)
	Change from baseline[c], mean (SD)	0.2 (1.26)	0.0 (0.62)
Week 8[b]	n	291	69
	Mean (SD)	0.3 (1.39)	0.3 (0.81)
	Change from baseline[c], mean (SD)	0.2 (1.43)	0.1 (0.91)
Week 12[b]	n	287	73
	Mean (SD)	0.2 (0.89)	0.2 (0.90)
	Change from baseline[c], mean (SD)	0.1 (0.93)	0.1 (0.78)
Week 24[b]	n	261	70
	Mean (SD)	0.2 (0.75)	0.1 (0.55)
	Change from baseline[c], mean (SD)	0.1 (0.88)	0.0 (0.79)
Week 36[b]	n	—	69
	Mean (SD)		0.1 (0.56)
	Change from baseline[c], mean (SD)		-0.1 (0.68)
Week 52[b]	n	—	68
	Mean (SD)		0.0 (0.24)
	Change from baseline[c], mean (SD)		-0.1 (0.61)

[a]Baseline was defined as the last non-missing observation prior to the date of the patient's enrollment in the study on Day 0; [b]Post-baseline only included data from patients who started the first investigational product on/before the visit evaluated; [c]For change from baseline summaries, only patients with a value at both baseline and a post-baseline visit were included.

FSS = full safety set; QSS-NM = Quantitative Scoring Scale for Evaluation of the Nasal Mucosa; SD = standard deviation.

Table 3. Overall Interpretation of Upper Nasal Endoscopy by Visit

Period		24-week FSS (N=354)	52-week FSS (N=73)
Baseline[a], n (%)	n	354	73
	Normal	328 (92.7)	68 (93.2)
	Abnormal, NCS	26 (7.3)	5 (6.8)
	Abnormal, CS	0	0
Week 4[b], n (%)	n	308	71
	Normal	283 (91.9)	65 (91.5)
	Abnormal, NCS	21 (6.8)	6 (8.5)
	Abnormal, CS	4 (1.3)	0
Week 8[b], n (%)	n	291	69
	Normal	275 (94.5)	65 (94.2)
	Abnormal, NCS	13 (4.5)	3 (4.3)
	Abnormal, CS	3 (1.0)	1 (1.4)
Week 12[b], n (%)	n	287	73
	Normal	268 (93.4)	68 (93.2)
	Abnormal, NCS	16 (5.6)	4 (5.5)
	Abnormal, CS	3 (1.0)	1 (1.4)
Week 24[b], n (%)	n	261	70
	Normal	248 (95.0)	68 (97.1)
	Abnormal, NCS	13 (5.0)	2 (2.9)
	Abnormal, CS	0	0
Week 36[b], n (%)	n	—	69
	Normal		65 (94.2)
	Abnormal, NCS		4 (5.8)
	Abnormal, CS		0
Week 52[b], n (%)	n	—	68
	Normal		64 (94.1)
	Abnormal, NCS		4 (5.9)
	Abnormal, CS		0

[a]Baseline was defined as the last non-missing observation prior to the date of the patient's enrollment in the study on Day 0; [b]Post-baseline only included data from patients who started the first INP104 dose on/before the visit evaluated.

CS = clinically significant; FSS = full safety set; NCS = not clinically significant.

Figure 2. Nasal TEAEs Associated With Nasal Endoscopy Findings and UPSIT Scores (24-Week FSS, N=354)

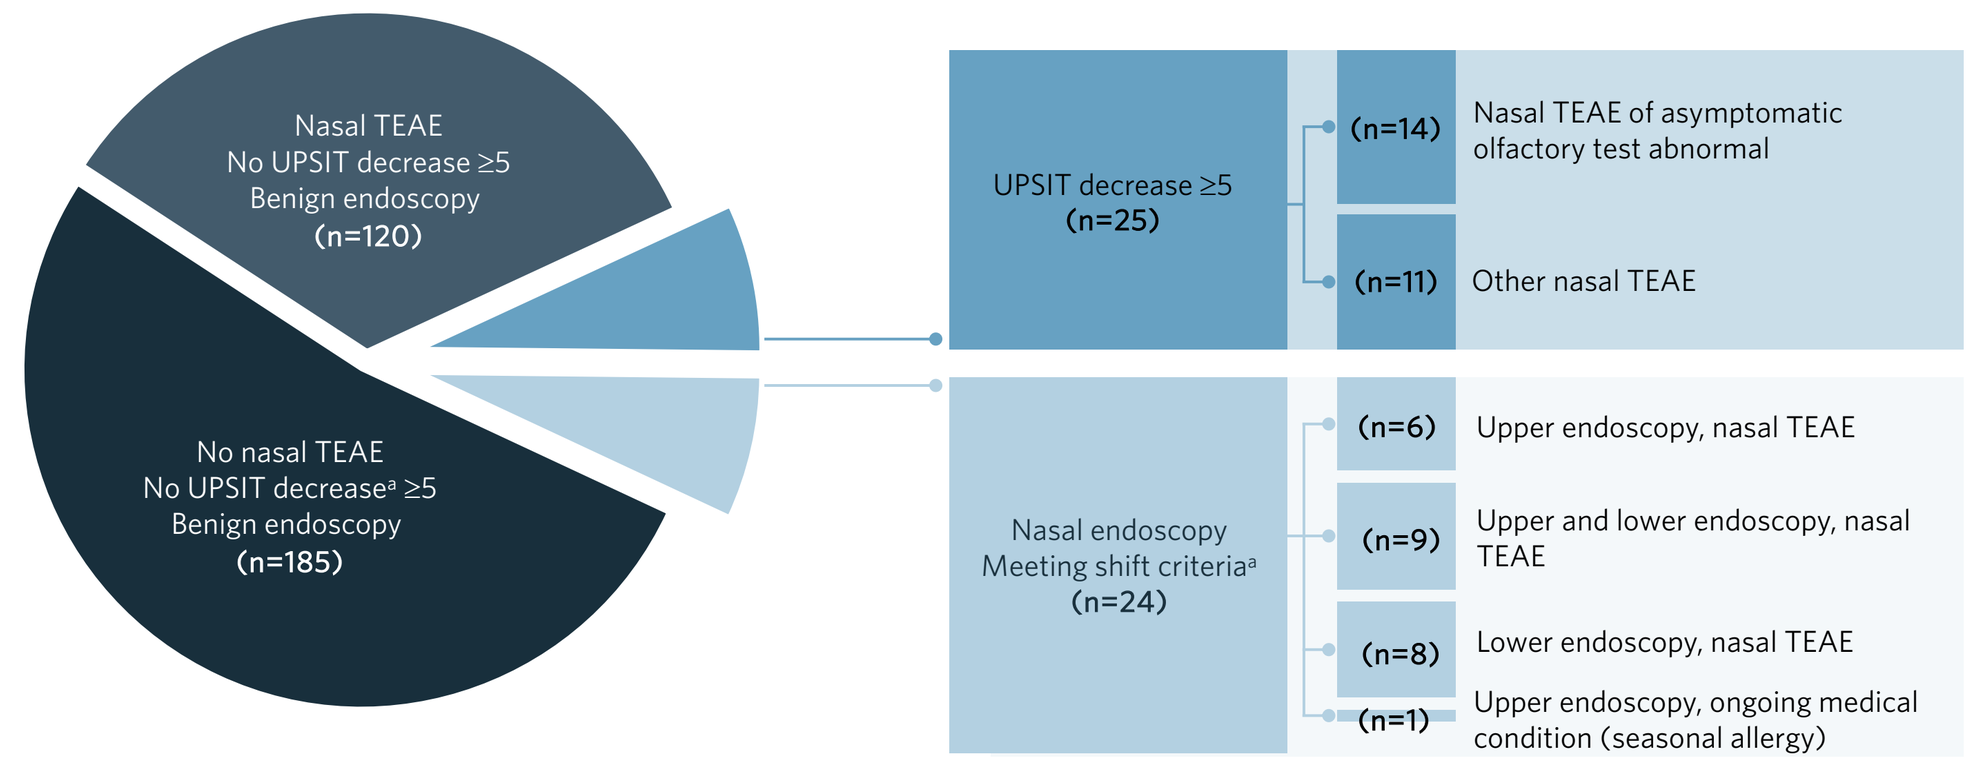
Nasal TEAE
No UPSIT decrease ≥5
Benign endoscopy
(n=120)
No nasal TEAE
No UPSIT decrease[a] ≥5
Benign endoscopy
(n=185)
UPSIT decrease ≥5
(n=25)
(n=14) Nasal TEAE of asymptomatic olfactory test abnormal
(n=11) Other nasal TEAE
Nasal endoscopy
Meeting shift criteria[a]
(n=24)
(n=6) Upper endoscopy, nasal TEAE
(n=9) Upper and lower endoscopy, nasal TEAE
(n=8) Lower endoscopy, nasal TEAE
(n=1) Upper endoscopy, ongoing medical condition (seasonal allergy)

[a]Upper endoscopy shift criteria include a total upper QSS-NM score >7, an individual upper score ≥2, or an overall impression of "abnormal clinically significant." Lower endoscopy shift criteria include an overall impression of "abnormal clinically significant."

FSS = full safety set; QSS-NM = Quantitative Scoring Scale for Evaluation of the Nasal Mucosa; TEAE = treatment-emergent adverse event; UPSIT = University of Pennsylvania Smell Identification Test.

Conclusion

- Mild-to-moderate mucosal edema was observed in 13 patients (5.0%) on endoscopy but was not considered a concerning finding
- The UPSIT detected 17 cases of subclinical instances of olfactory reduction in patients with nasal TEAEs, most of which resolved (14 cases)
- Nasal AE monitoring revealed that a small percentage of patients experienced nasal discomfort and congestion leading to treatment discontinuation in the 24-week period (1.1% and 1.4%, respectively)
- In the independent opinion of the NSRC:
 - Patient-reported AEs were sufficient to monitor safety of intermittent use of INP104
 - Nasal endoscopies and associated QSS-NM scoring and physiological tests did not add additional clinical value
 - No safety concerns were raised based on review of all data

References

1. Hoekman J, et al. *US Neurol.* 2020;16:25–31.
2. Shrewsbury SB, et al. *Headache.* 2019;59:394–409.
3. Aurora S, et al. *Neurology.* 2021;96(Suppl 15).2490.
4. Doty RL, et al. *Laryngoscope.* 1984;94:176-178.
5. Psaltis AJ, et al. *The Laryngoscope.* 2014;124:2216-2223.

Disclosures and Acknowledgments

Greg Davis is a consultant for Impel NeuroPharma, AstraZeneca, Intersect ENT, Medtronic, Stryker, OptiNose, and Smith & Nephew. Adil Fatakia consults for Aerin Medical Inc., Medtronic, Intersect ENT, and Tandem Clinical Research. Seth Pransky consults for Inovio, Otonomy, and PhotoniCare. Sheena K. Aurora, Jasna Hocevar-Trnka, and Stephen B. Shrewsbury are full-time employees of Impel NeuroPharma and are stockholders in Impel NeuroPharma. Stephen B. Shrewsbury is an officer of Impel NeuroPharma. Alison E. Heald is a paid consultant to Impel NeuroPharma. This research was sponsored by Impel NeuroPharma. Editorial support was provided by IMPRINT Science and funded by Impel NeuroPharma. IMPEL, POD, and the IMPEL Logo are registered trademarks of Impel NeuroPharma.

Impel NEUROPHARMA®

Presented at the Virtual AHS Annual Scientific Meeting, June 3–6, 2021