Recurrence Rates for INP104 for the Acute Treatment of Migraine: Results From the Phase 3 STOP 301 Study

Stewart J. Tepper, MD[1*]; Jessica Ailani, MD[2]; Stephen B. Shrewsbury, MB ChB[3]; Sheena K. Aurora, MD[3]

[1]Geisel School of Medicine at Dartmouth, Hanover, NH, USA; [2]MedStar Georgetown University Hospital, Washington, DC, USA; [3]Impel NeuroPharma, Seattle, WA, USA

*Presenting author

Introduction

- Recurrence of migraine headache is common with acute therapies, particularly with the triptan class[1-4]
- In some patients, recurrence of migraine may lead to an increased risk of new-onset chronic migraine and/or development of medication overuse headache due to repeat dosing of acute medications[5-7]
- Importantly, repeat dosing can impact cost[5]; and data on the efficacy and/or safety of repeat dosing are scarce
- Despite advances in the acute care of migraine, recurrence is still an important issue for physicians and their patients, and new therapies are needed to address this unmet need[1,2]
- To determine the sustained or prolonged efficacy of an acute therapy, varying study endpoints are often set by clinical trials, and can include return of migraine following pain relief or freedom; use of rescue medication; or sustained pain freedom or relief at various timepoints[1-3,8,9]
- Intravenous (IV) dihydroergotamine mesylate (DHE) has a long history as an efficacious acute therapy for migraine patients, including those who experience migraine recurrence[10,11]
- INP104 is an investigational, novel drug-device product that targets delivery of DHE to the upper nasal space using Precision Olfactory Delivery (POD®) technology (**Figure 1**)[12]
- Exploratory efficacy data for the first INP104-treated migraine from the recent Phase 3, STOP 301 study have been previously presented[13]
 - Pain freedom was self-reported in 38.0% of patients, most bothersome symptom (MBS) freedom in 52.1%, and pain relief in 66.3% at 2 hours for the first INP104-treated migraine[13]
 - Pain and MBS freedom were not very different whether patients administered INP104 within 2 hours of migraine initiation or beyond for their first treated migraine[13]

Figure 1. (A) INP104 Product and (B) Actuation of INP104

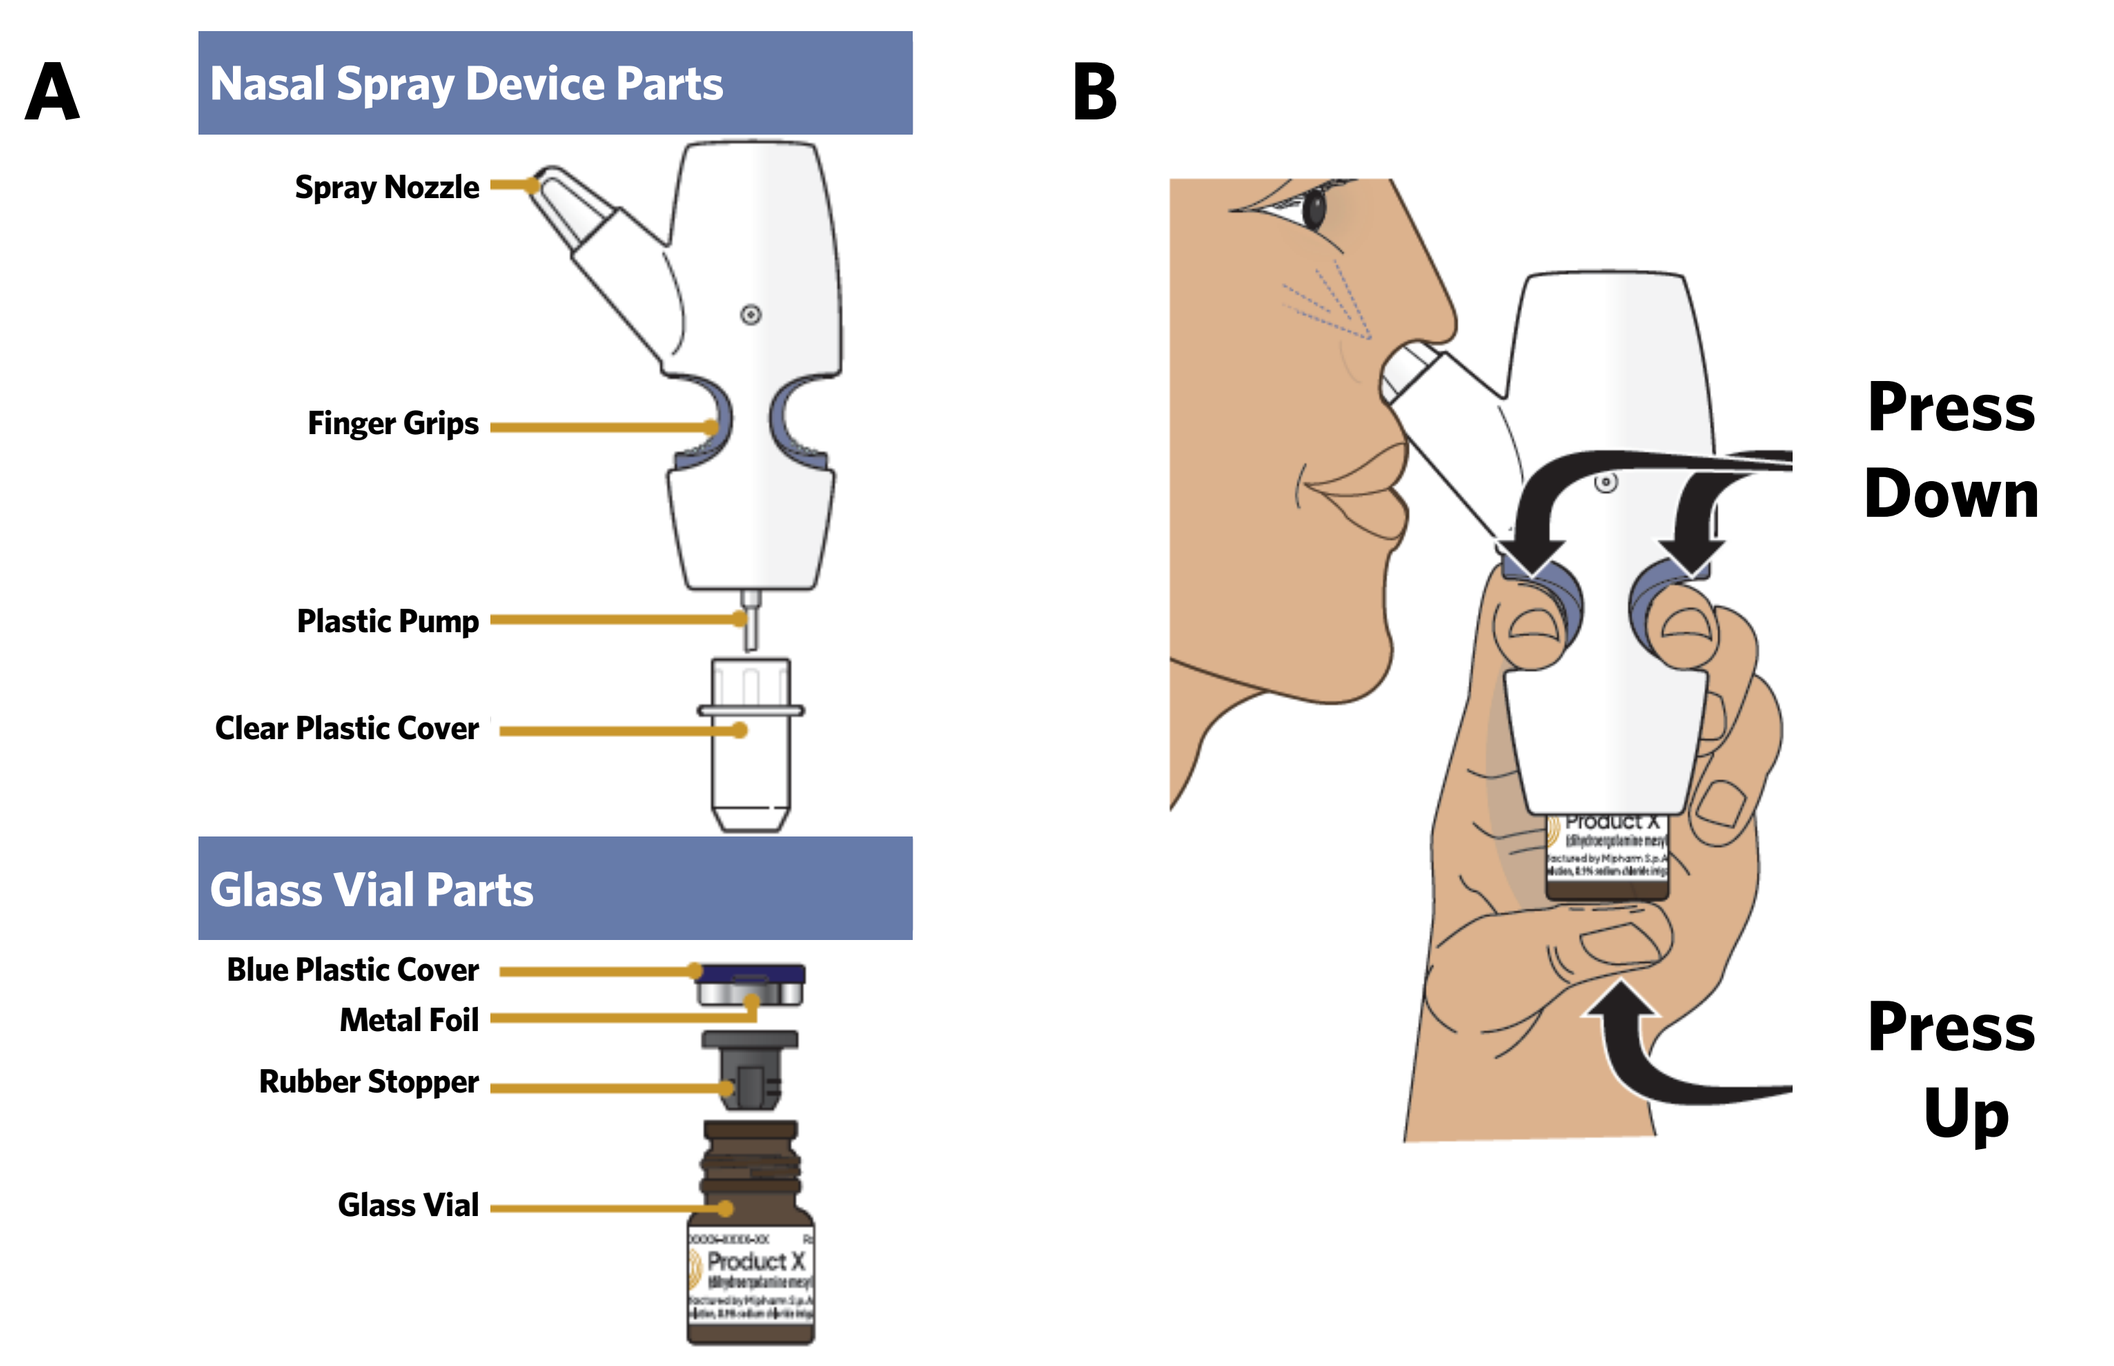
A
Nasal Spray Device Parts
Spray Nozzle
Finger Grips
Plastic Pump
Clear Plastic Cover
Glass Vial Parts
Blue Plastic Cover
Metal Foil
Rubber Stopper
Glass Vial
Product X
B
Press Down
Press Up

Objective

- Here we report recurrence rates; sustained pain freedom; and use of rescue medication for the first INP104-treated migraine to demonstrate single-dose efficacy associated with INP104 use from the STOP 301 study

Methods

Study Design

- STOP 301 was a Phase 3, interventional, open-label, single-group assignment study, assessing the safety, tolerability, and exploratory efficacy of INP104 over 24 and 52 weeks (NCT03557333)
- The study included a:
 - 4-week screening period, in which patients used their best usual care
 - 24-week treatment period for all patients, in which patients used INP104
 - Treatment extension to 52 weeks for a subset of the patients
 - 2-week post-treatment follow-up period
- After completing the screening period, all eligible patients were provided with up to 3 doses/week of INP104 to nasally self-administer (1.45 mg in a dose of 2 sprays) with self-recognized migraine attacks for either 24 and/or 52 weeks
- Patients completed daily eDiaries to capture headache and migraine details, headache medication usage, and MBS severity

Study Patients

- Patients were adult (18–65 years) males or females who had a documented diagnosis of migraine, with a minimum of 2 migraine attacks, with or without aura, each month not qualifying as chronic migraine during the previous 6 months per the *International Classification of Headache Disorders,* version 3 beta (ICHD-3β)
- Patients were in general good health, with no significant medical history or clinical abnormalities at baseline, which included no history of cardiovascular events

Study Outcome Measures

- Post hoc analyses of STOP 301 data were performed on self-reported exploratory efficacy data collected over 24 weeks
- Outcomes are reported for the 24-week full safety set (FSS; i.e., all patients who were enrolled and received ≥1 dose of INP104), which included 354 patients
- The eDiary did not capture 24- and 48-hour pain measurements if a patient was pain-free at 2 hours; therefore:
 - *Recurrence was defined as the percentage of patients who self-reported an onset of a new headache prior to 24 or 48 hours post-INP104 in patients who had self-reported pain freedom at 2 hours post-INP104*
 - *Sustained pain freedom was defined as the percentage of patients who self-reported no other headache at 24 and 48 hours post-INP104 in patients who had self-reported pain freedom at 2 hours post-INP104*
- After baseline, only non-ergot, non-triptan acute treatments were allowed as rescue medication for patients who still had headache pain, and only after 2 hours had elapsed since INP104 administration. A second dose of INP104 could also be used within 24 hours. The use of such rescue medications was recorded in the eDiary
 - *Rescue medication use was defined as the percentage of patients who self-reported using rescue medication following their first INP104-treated migraine*
 - Data included all treated migraine attacks regardless of availability of 2-hour pain and MBS results

Results

Recurrence

- Recurrence was self-reported in 7.1% (9/126) and 14.3% (18/126) of patients at 24 hours and 48 hours post-INP104, respectively, for the first treated migraine (**Figure 2**)

Figure 2. Recurrence Rates for the First INP104-Treated Migraine

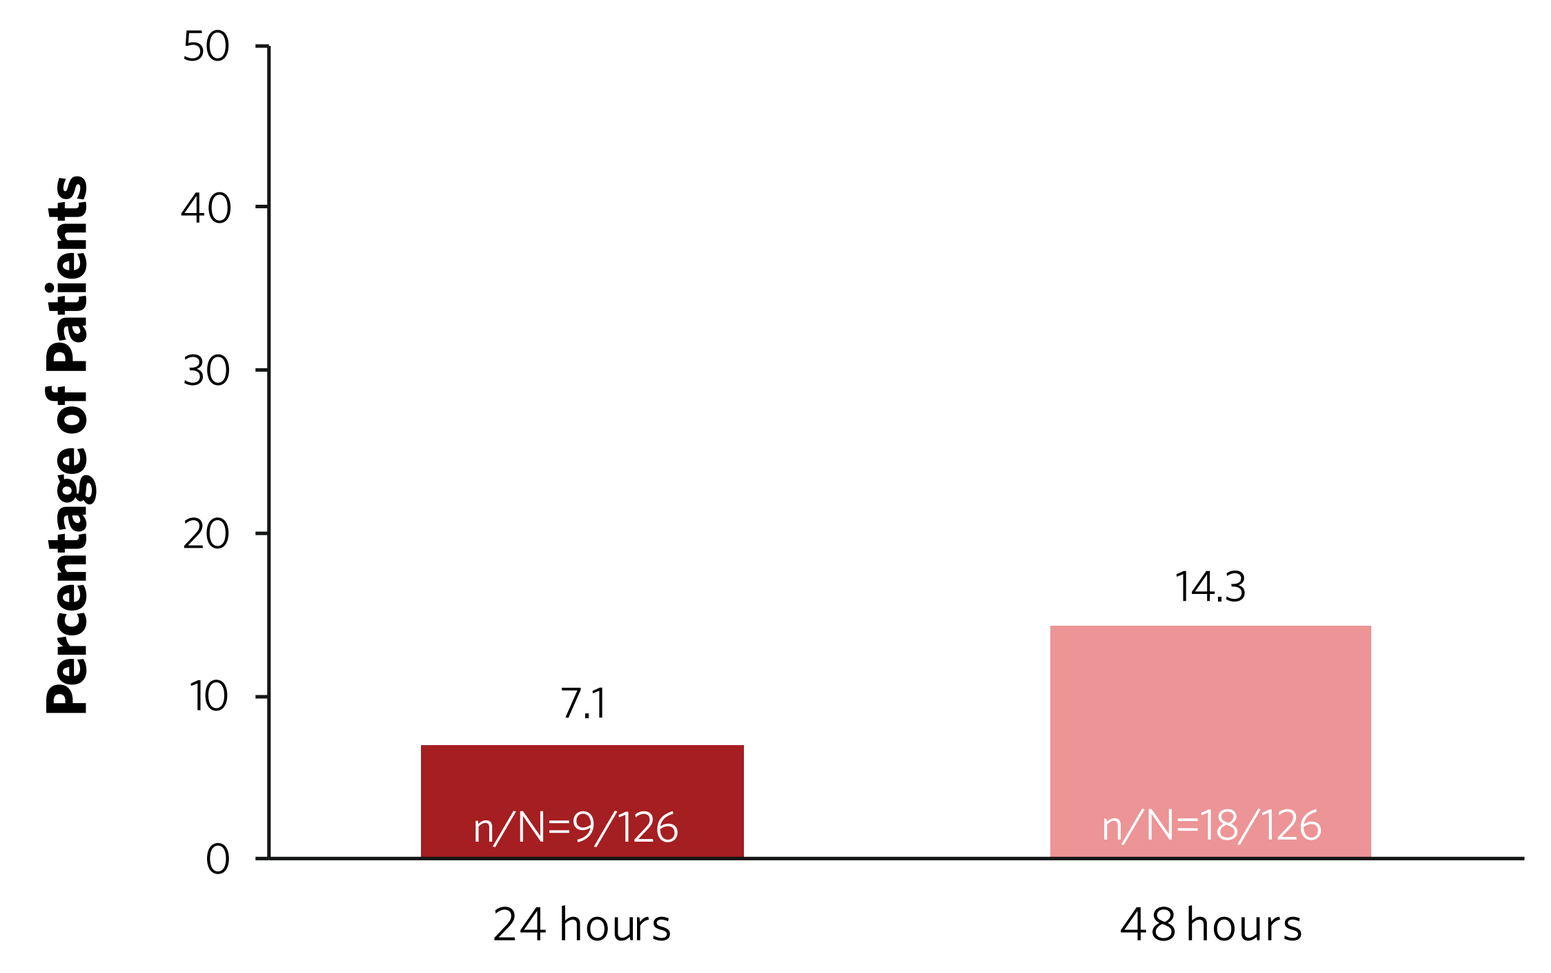
Percentage of Patients
50
40
30
20
10
0
7.1
n/N=9/126
14.3
n/N=18/126
24 hours
48 hours

Note: Data are self-reported.

Sustained Pain Freedom

- Pain freedom was self-reported by 38% (126/332) of patients at 2 hours after treating their first migraine with INP104
- Sustained pain freedom was self-reported by 35.2% (117/332) and 32.5% (108/332) of patients at 24 and 48 hours post-INP104, respectively, for the first treated migraine (**Figure 3**)

Figure 3. Pain- and Sustained-Pain Freedom Rates for the First INP104-Treated Migraine

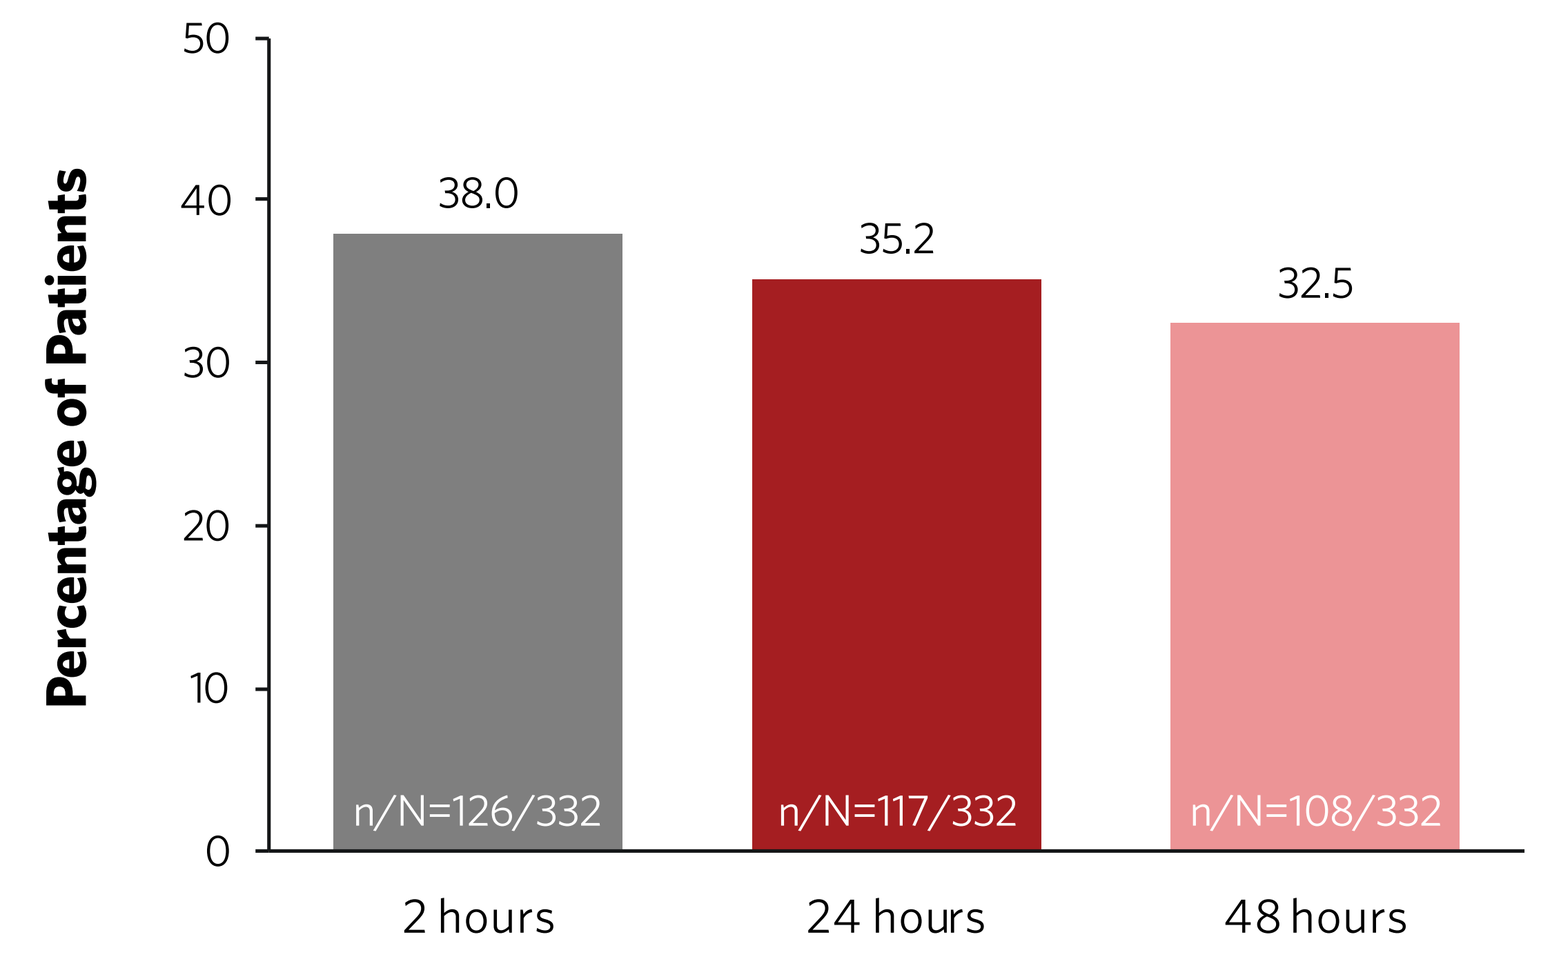
Percentage of Patients
50
40
30
20
10
0
38.0
n/N=126/332
35.2
n/N=117/332
32.5
n/N=108/332
2 hours
24 hours
48 hours

Note: Data are self-reported.

Rescue Medication

- Rescue medication use was self-reported by 18.2% (63/347) of patients for the first INP104-treated migraine (**Figure 4**)

Figure 4. Rescue Medication Use for the First INP104-Treated Migraine

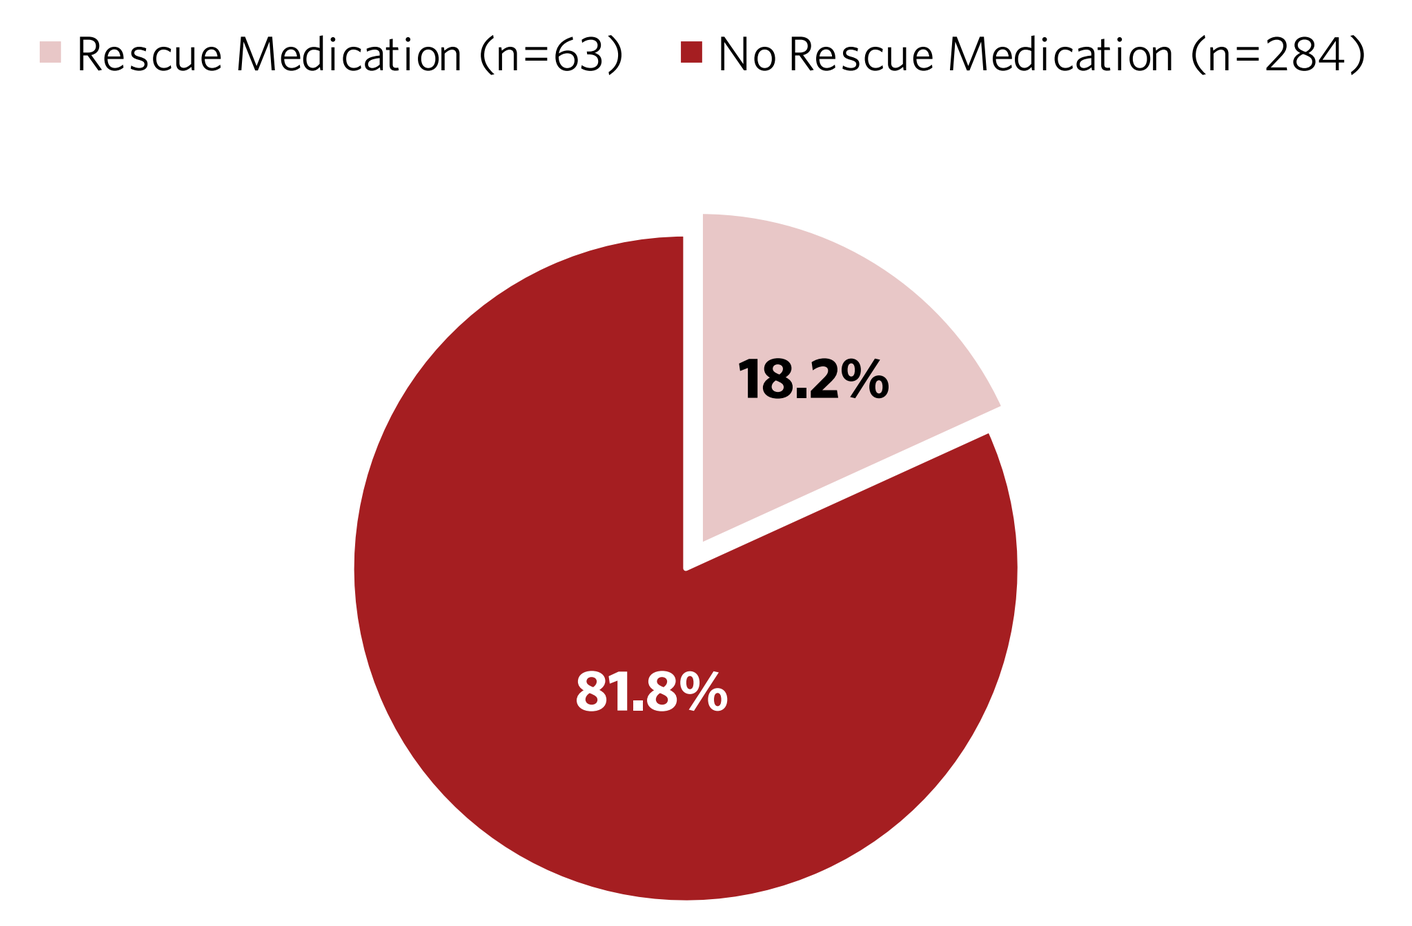
Rescue Medication (n=63)
No Rescue Medication (n=284)
18.2%
81.8%

Note: Data are self-reported.

Conclusion

- A post hoc analysis of exploratory efficacy data from the STOP 301 study was performed to evaluate single-dose efficacy of INP104 for the first treated migraine
 - Recurrence rates associated with INP104 were low; 7.1% and 14.3% at 24 hours and 48 hours, respectively
 - Sustained pain freedom was self-reported in 35.2% and 32.5% of patients at 24 and 48 hours post-INP104, respectively
 - 18.2% of patients self-reported using rescue medication post-INP104
- Data from this Phase 3 study demonstrate that a single dose of INP104 was associated with high rates of sustained efficacy for those who had received relief with INP104 for their first treated migraine
 - Results also align with previously presented data showing sustained efficacy of INP104 over multiple migraine attacks[13]

References

1. Aurora SK, Dempsey J. *Curr Treat Options Neurol.* 2002;4:335-342.
2. Diener HC, et al. *Cephalalgia.* 2019;39:687-710.
3. Ferrari MD, et al. *Lancet.* 2001;358:1668-1675.
4. Ryan R, et al. *Headache.* 2002;42(Suppl 2):S84-92.
5. Atlas S, et al. Acute treatments for migraine: effectiveness and value. Institute for Clinical and Economic Review, February 25, 2020.
6. Lipton RB, et al. *Neurology.* 2015;84:688-695.
7. Ferrari MD, et al. *Cephalalgia.* 2002;22:633-658.
8. Croop R, et al. *Lancet.* 2019;394:737-745.
9. Brandes JL, et al. *Cephalalgia.* 2019;39:1343-1357.
10. Silberstein SD, et al. *Headache.* 2020;60:40-57.
11. Morren JA, Galvez-Jimenez N. *Expert Opin Pharmacother.* 2010;11:3085-3093.
12. Shrewsbury SB, et al. *Headache.* 2019;59:394-409.
13. Aurora S, et al. *Neurology.* 2021;96(Suppl 15).2490.

Disclosures and Acknowledgments

Stewart J. Tepper has received grants for research (no personal compensation) from Allergan, Amgen, Eli Lilly and Company, Lundbeck, Neurolief, Novartis, Satsuma, and Zosano. He has consulted and/or participated in Advisory Boards (honoraria) for Aeon, Align Strategies, Allergan/AbbVie, AlphaSights, Amgen, Aperture Venture Partners, Aralez Pharmaceuticals Canada, Axsome Therapeutics, Becker Pharmaceuticals, BioDelivery Sciences International, Biohaven, ClearView Healthcare Partners, CRG, Currax, Decision Resources Group, DeepBench, Eli Lilly, Equinox, ExpertConnect, GLG, Guidepoint, Healthcare Consultancy Group, Health Science Communications, HMP Communications, Impel, Krog and Partners, Lundbeck, M3 Global Research, Magellan Rx Management, Medicxi, Navigant Consulting, Neurolief, Nordic BioTech Advisors ApS, Novartis, Pulmatrix, Reckner Healthcare, Relevale, SAI MedPartners, Satsuma, Slingshot Insights, Spherix Global Insights, Sudler and Hennessey, Synapse Medical Communications, System Analytic, Teva, Theranica, Thought Leader Select, Trinity Partners, Xoc, and Zosano. He receives a salary from Dartmouth-Hitchcock Medical Center, American Headache Society, and Thomas Jefferson University. He has received CME honoraria from the American Academy of Neurology; American Headache Society; Cleveland Clinic Foundation; Diamond Headache Clinic; Elsevier; Forefront Collaborative; Hamilton General Hospital, Ontario, Canada; Headache Cooperative of New England; Henry Ford Hospital, Detroit; Inova; Medical Learning Institute; Peerview; Medical Education Speakers Network; Miller Medical Communications; North American Center for CME; Physicians' Education Resource; Rockpointe; ScientiaCME; and WebMD/Medscape. Jessica Ailani has received honoraria for consulting from Amgen, AbbVie, Biohaven, Eli Lilly and Company, Lundbeck, Teva, Impel, Satsuma, Axsome, and Vorso. She has received stock options from Ctrl M Health for consulting. She has received honoraria for speaking engagements from AbbVie, Amgen, Biohaven, Eli Lilly, Lundbeck, and Teva. Jessica Ailani's institution has received funding for clinical trials for her work as principal investigator from Allergan/AbbVie, Biohaven, Eli Lilly and Company, Satsuma, and Zosano. She has provided advising and editorial services and received honoraria from Current Pain and Headache Reports, SELF, Neurology Live, and Medscape. Sheena K. Aurora and Stephen B. Shrewsbury are full-time employees of Impel NeuroPharma and are stockholders in Impel NeuroPharma. Stephen B. Shrewsbury is an officer of Impel NeuroPharma. This research was sponsored by Impel NeuroPharma. Editorial support was provided by IMPRINT Science and funded by Impel NeuroPharma. IMPEL, POD, and the IMPEL Logo are registered trademarks of Impel NeuroPharma.

Impel NEUROPHARMA®

Presented at the Virtual AHS Annual Scientific Meeting, June 3–6, 2021