Real-World Assessment of Baseline Demographic and Clinical Characteristics Among Patients Using INP104 in the United States

Trupti Gokani, MD[1]; Linda Nguyen, MD[2]; Robert E. Vann, PhD[3]; Judie Gutierrez, PhD, MBA[3]; Sutapa Ray, PhD[3]; Sheena K. Aurora, MD[3]

[1]Neura Health, Virtual Clinic, USA; [2]Stanford University, Stanford, CA, USA; [3]Impel Pharmaceuticals, Seattle, WA, USA

Introduction

- INP104 is a novel drug-device combination product that delivers dihydroergotamine mesylate (DHE) to the upper nasal space using Precision Olfactory Delivery (POD®) and was approved by the Food and Drug Administration for the acute treatment of migraine in September 2021[1,2]
- INP104 offers a non oral, non-invasive, and convenient alternative to DHE administered intravenously, (which can provide rapid symptom relief but can often cause nausea and vomiting and must be administered in a clinical setting) as well as superior pharmacokinetics compared to MIGRANAL (the first approved DHE mesylate nasal spray)[1,3-5]
- The long-term safety and exploratory efficacy of INP104 in patients with migraine has been demonstrated in a Phase 3, open-label, study (STOP 301), which has been previously published[1]
- Real-world data are needed to understand characteristics and medication utilization patterns of patients who use INP104 to determine how it fits into treatment patterns for migraine relief

Objective

- To report the baseline demographic and clinical characteristics among patients who used INP104 over a 12-month period in a real-world setting

Methods

- This was a retrospective, longitudinal, observational study designed to assess demographic and clinical characteristics, identify baseline comorbidities and concomitant medication use, and assess treatment patterns among patients with migraine who were treated with INP104 based on medical and pharmacy claims in the United States from the STATinMED database
- Eligibility criteria (Figure 1, Figure 2)
 - Diagnosis of migraine during the study period spanning January 1, 2020, through October 31, 2022
 - ≥18 years of age at index
 - ≥1 pharmacy claim for INP104 use during a patient identification period spanning October 1, 2021, through October 31, 2022
 - Continuous enrollment with both medical and pharmacy benefits for 12 months pre- and post-index period
 - No diagnoses of malignant neoplasm at any body location (particularly the central nervous system) at any time during the study period
- The baseline period was defined as the 12 months prior to the index period (not including the index period)
- Clinical characteristics:
 - Quan-Charlson Comorbidity Index (CCI) score during baseline period: The most used index in health outcomes studies is the CCI, which assigns a weight ranging from 1 to 6 according to disease severity for 19 conditions. The index has since been revised; the Quan-CCI, which assesses 17 comorbidities, was used for this study. Quan-CCI score was assessed for the baseline period and can also be categorized as 0, 1, 2, 3, 4, and 5+ as a proxy for baseline severity
 - Key baseline comorbidities during the baseline period: A binary variable was created using ICD-9/ICD-10-CM codes for patients diagnosed with the following comorbidities commonly associated with migraine:
 - Psychiatric disorders (depression, anxiety disorder, bipolar disorder, abuse, post-traumatic stress disorder)
 - Cardiovascular diseases (stroke, myocardial infarction, angina)
 - Other neurological conditions (not including epilepsy or seizure, migraine, other headache syndromes)
 - Asthma
 - Obesity
 - Sleep disorders (restless leg syndrome, obstructive sleep apnea, insomnia)
 - Autoimmune diseases (rheumatoid arthritis, hypothyroidism)
 - Gastrointestinal (GI) disorders (irritable bowel syndrome, gastric ulcer/GI bleeding, gastroesophageal reflux disease, *Helicobacter pylori* infection, gastroparesis, hepatobiliary disorders, celiac disease)
 - Other pain syndromes (fibromyalgia, neck pain, back pain)
 - Epilepsy

Figure 1. Study Design

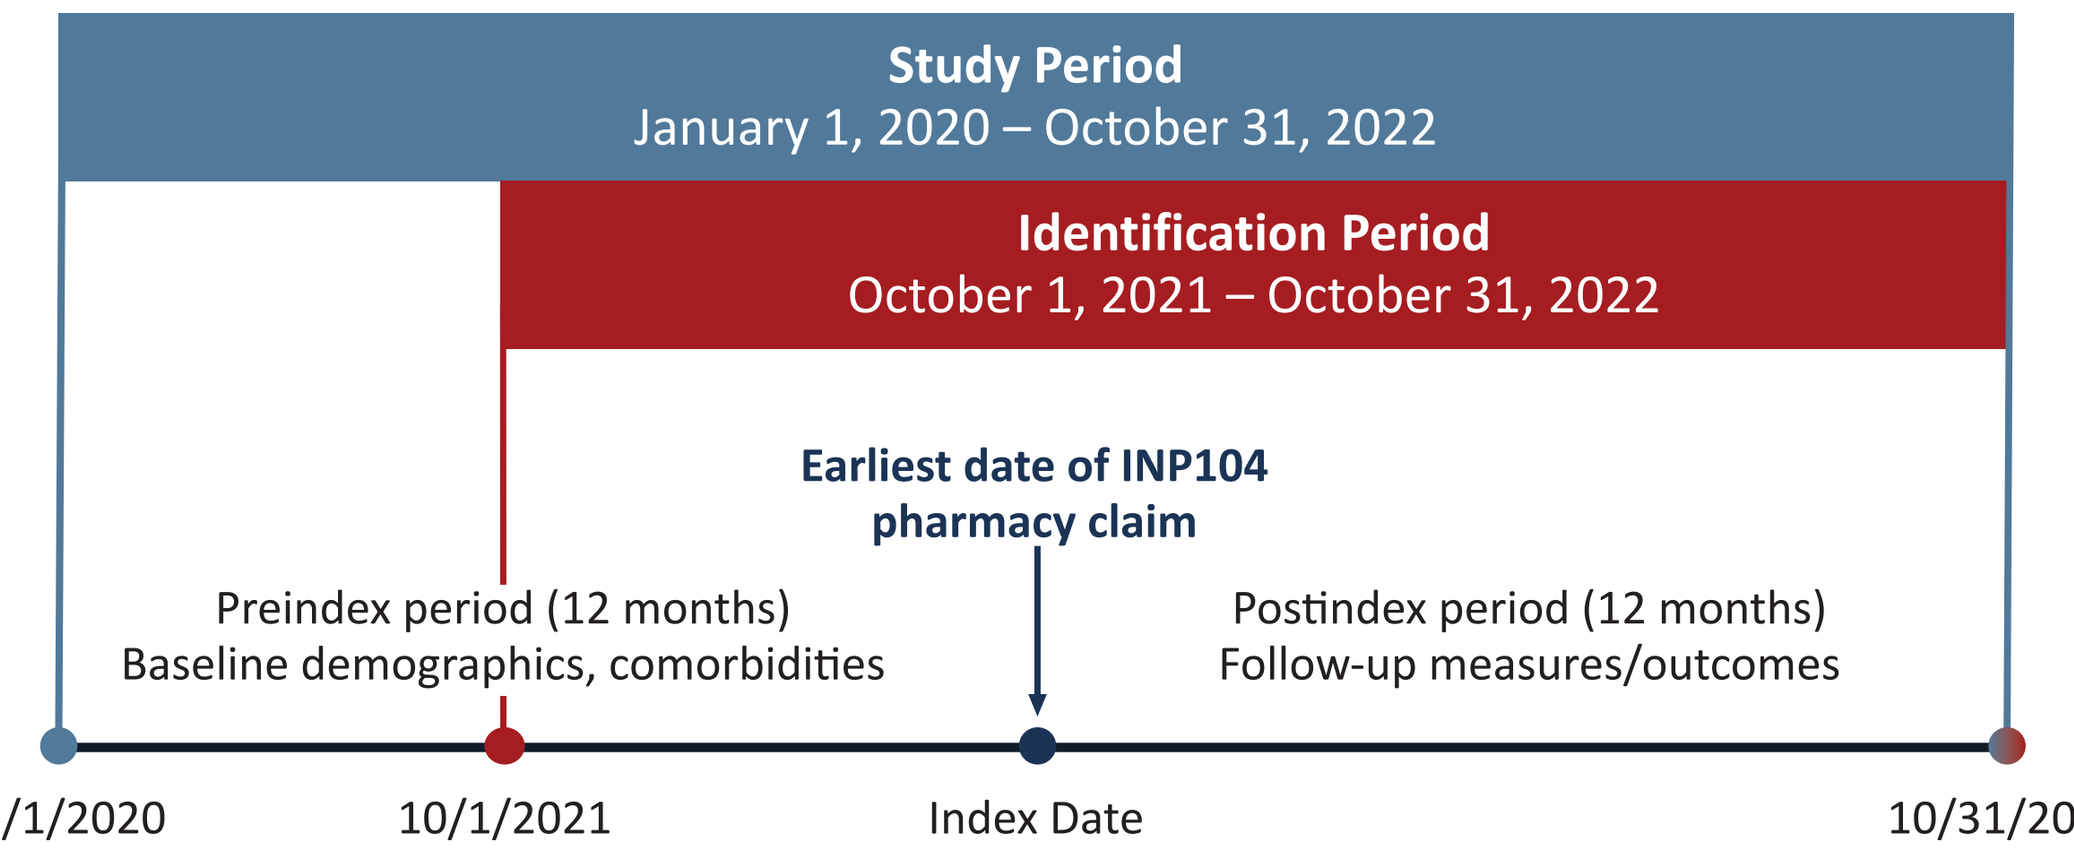
Study Period
January 1, 2020 – October 31, 2022
Identification Period
October 1, 2021 – October 31, 2022
Earliest date of INP104 pharmacy claim
Preindex period (12 months)
Baseline demographics, comorbidities
Postindex period (12 months)
Follow-up measures/outcomes
01/1/2020
10/1/2021
Index Date
10/31/2022

Figure 2. Patient Disposition

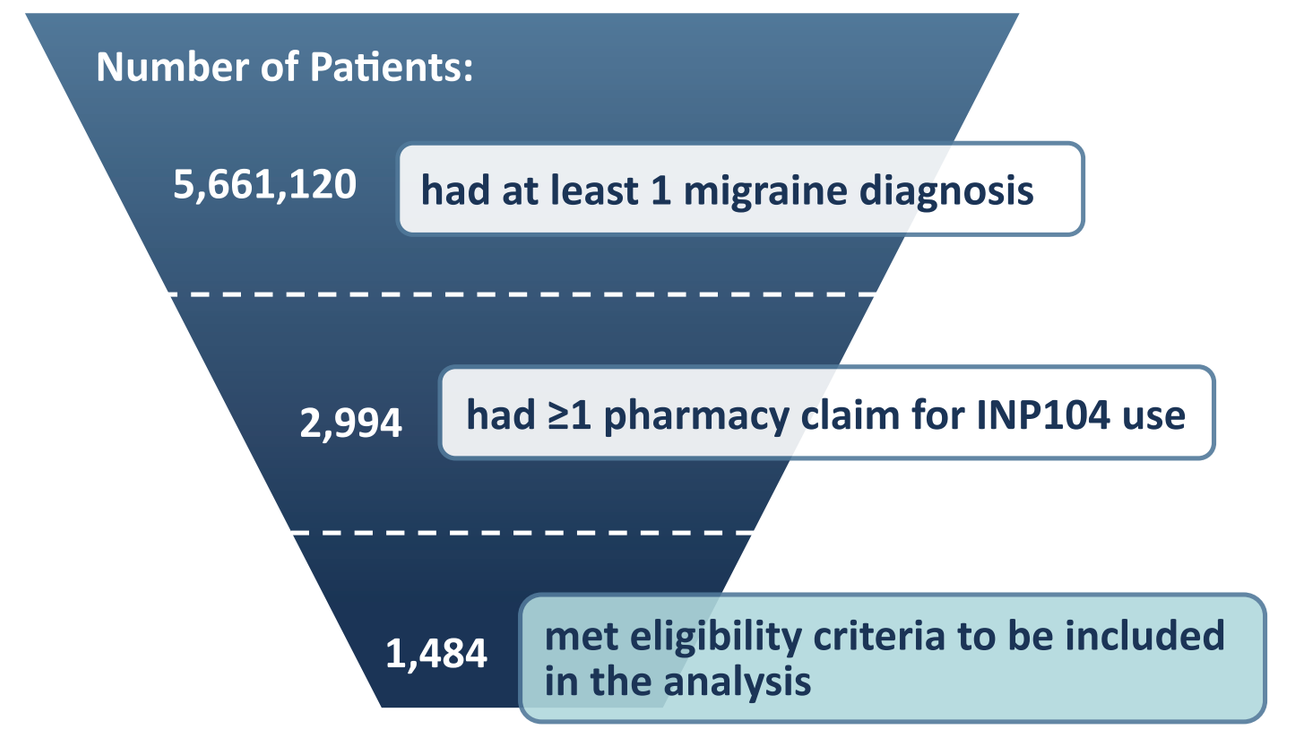
Number of Patients:
5,661,120
had at least 1 migraine diagnosis
2,994
had ≥1 pharmacy claim for INP104 use
1,484
met eligibility criteria to be included in the analysis

Results

Patient Baseline and Clinical Characteristics

- Most patients were female (87.9%) with a mean age of 43.4 years (Figure 3)
- Regional distribution across the United States was mostly in the South (42.1%) (Figure 4)
- The payer channel was mostly commercial (86.5%; Figure 5)
- The mean (SD) Charlson Comorbidity Index score was 0.31 (0.83) (Table 1)
- The most common baseline comorbidities (>10%) included headache syndromes other than migraine (36.7%), other neurological conditions (25.9%), other pain disorders (24.8%), sleep disorders (18.8%), GI disorders (15.9%) and psychiatric disorders (11.5%; Figure 6, Table 2)

Table 1. Baseline Age and Sex Distribution

Baseline Characteristic	N=1484
Age, years	
Mean/SD	43.4 (11.8)
Median	44
Sex, %	
Female	87.9%
Male	12.1%

SD=standard deviation.

Figure 3. Baseline Age Distribution by Age Group

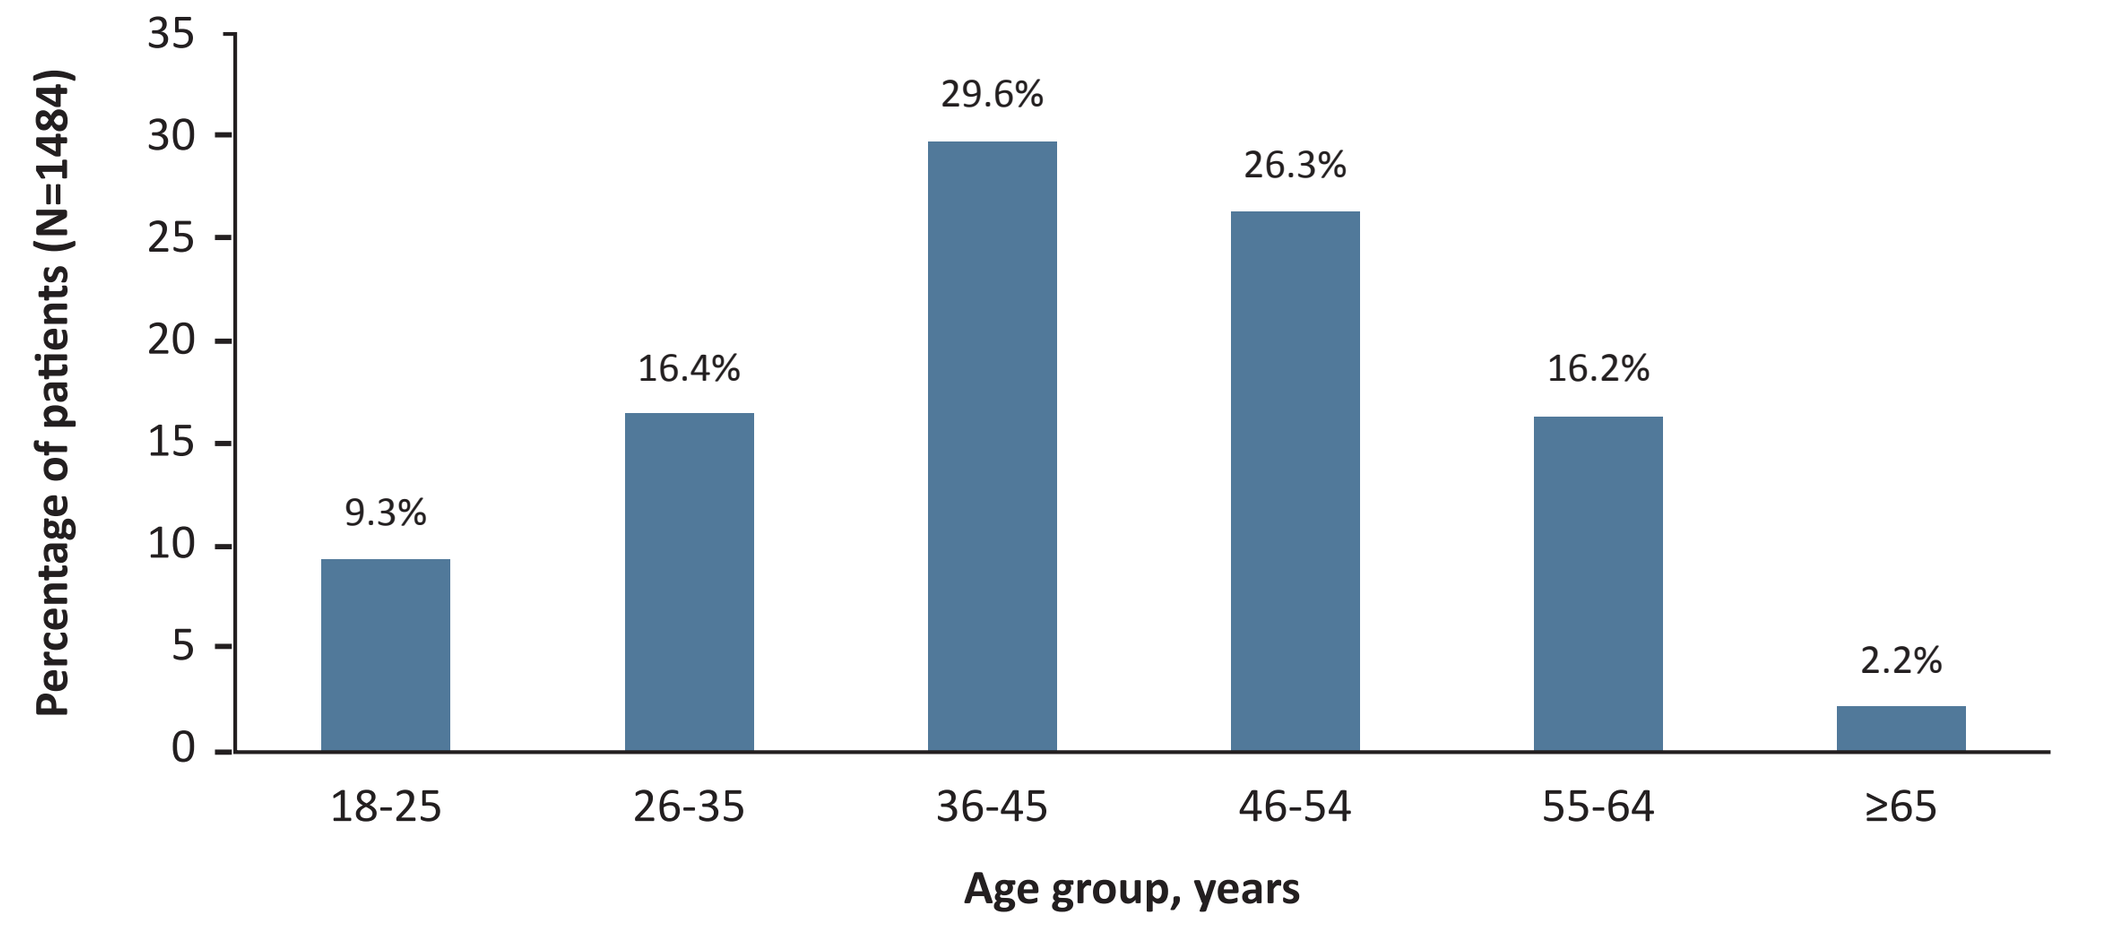
Percentage of patients (N=1484)
35
30
25
20
15
10
5
0
9.3%
16.4%
29.6%
26.3%
16.2%
2.2%
18-25
26-35
36-45
46-54
55-64
≥65
Age group, years

US=United States.

Figure 4. Baseline Patient Characteristics, Geographic Distribution

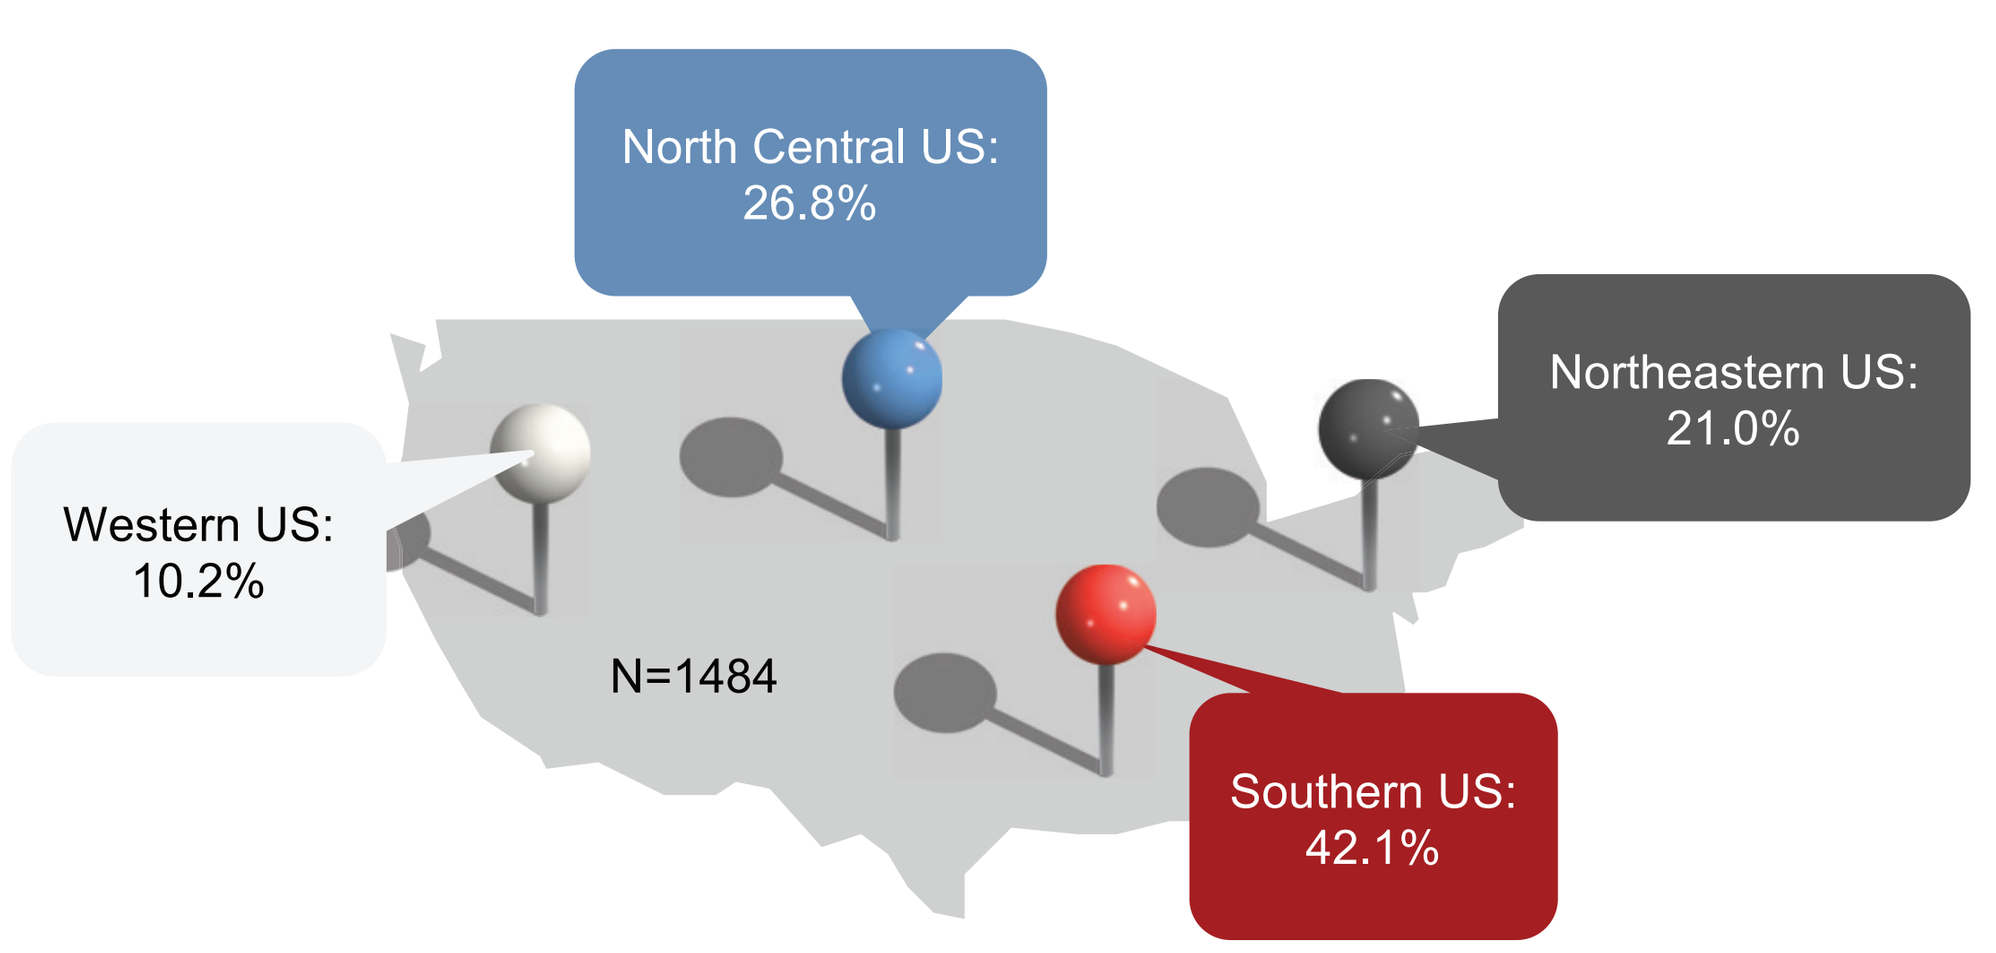
North Central US: 26.8%
Northeastern US: 21.0%
Western US: 10.2%
N=1484
Southern US: 42.1%

US=United States.

Figure 5. Baseline Patient Characteristics, Payer Channel

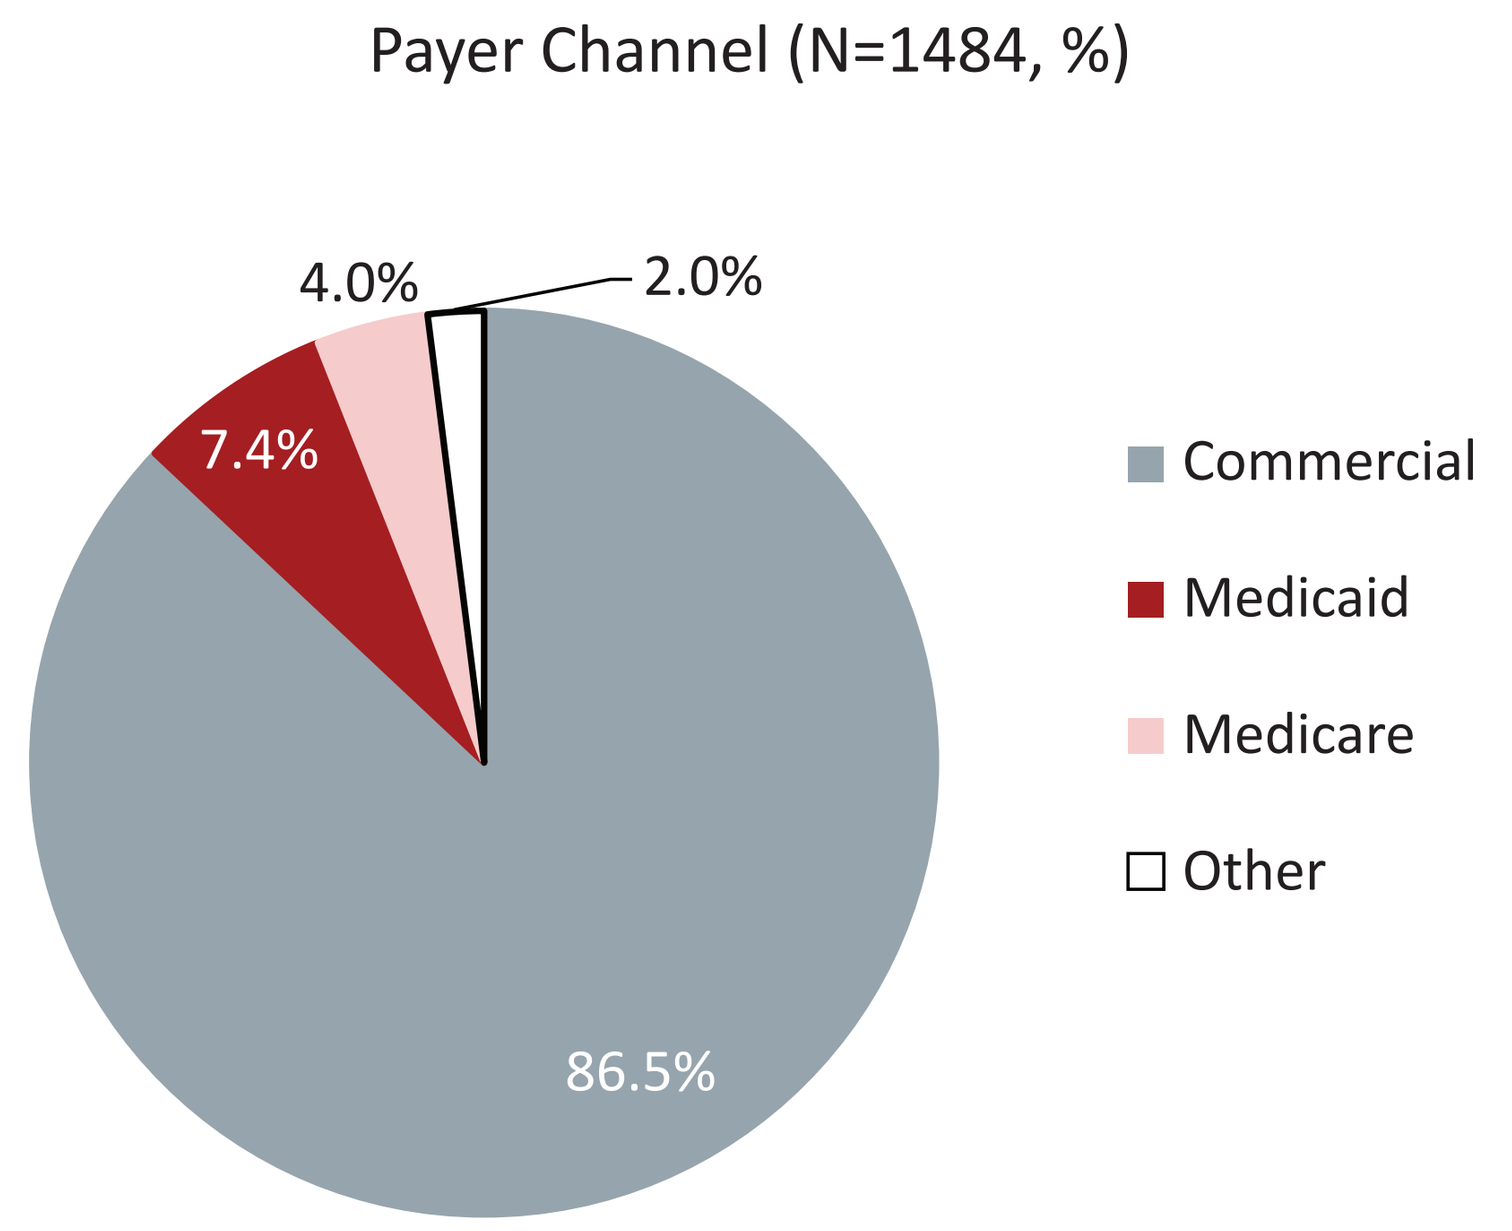
Payer Channel (N=1484, %)
4.0%
2.0%
7.4%
86.5%
Commercial
Medicaid
Medicare
Other

Figure 6. Headache Related Comorbidities Present at Baseline

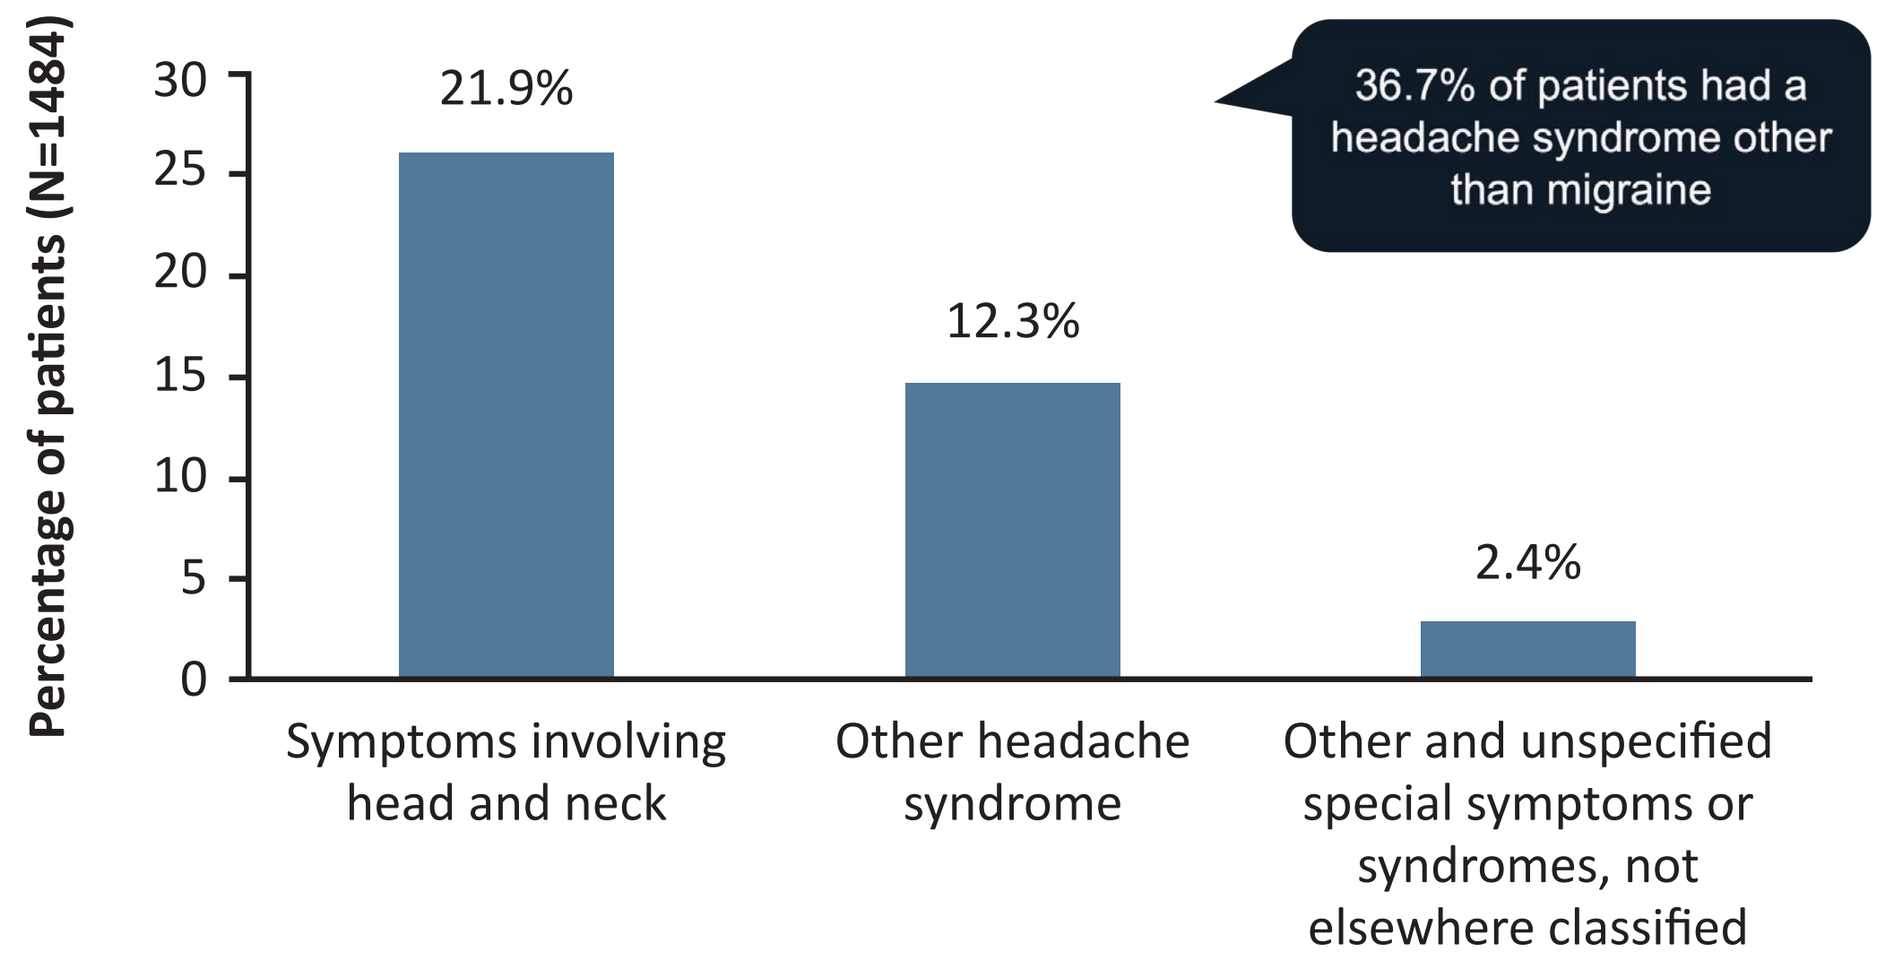
Percentage of patients (N=1484)
30
25
20
15
10
5
0
21.9%
12.3%
2.4%
36.7% of patients had a headache syndrome other than migraine
Symptoms involving head and neck
Other headache syndrome
Other and unspecified special symptoms or syndromes, not elsewhere classified

Table 2. Baseline Comorbidities

	Current Study N=1484	AMPP Studies[6,7] Episodic Migraine
Other neurological conditions	25.9%	--
Other pain disorders (fibromyalgia, neck pain, and back pain)	24.8%	~37%[c]
Sleep disorders	18.8%	--
Gastrointestinal disorders	15.9%[a]	8%[d]
Psychiatric disorders	11.5%[b]	48-64%[e]
Autoimmune diseases	8.9%	6.5%[f]
Obesity	8.2%	21%
Asthma	7.2%	~17%
Epilepsy or seizure	5.6%	2.6%[g]
Mononeuropathies of upper or lower limb	3.4%	--
Epilepsy	1.6%	--
Other and unspecified disorders of the nervous system	0.4%	--
Conversion disorder with seizures or convulsions	0.2%	--
Post-traumatic seizures	0%	--
Cardiovascular diseases	0.7%	6.3%[h]

[a]Includes irritable bowel syndrome (IBS), gastric ulcer/GI bleeding, gastroesophageal reflux disease (GERD), Helicobacter pylori infection, gastroparesis, hepatobiliary disorders, celiac disease). [b]Includes anxiety, bipolar disorder, depression, post-traumatic stress disorder, and substance abuse. [c]Chronic pain, rheumatoid arthritis pain. [d]Limited to ulcers of the stomach/intestines. [e]Includes anxiety, bipolar disorder, and depression. [f]Limited to dermatitis/eczema. [g]Included seizures, epilepsy, fits, or convulsions. [h]Limited to heart disease/angina. AMPP=American Migraine Prevalence and Prevention.

Conclusions

- Real-world evidence demonstrates that most patients with migraine who used INP104 as an acute therapy are females between the ages of 36 and 45 years, and have comorbidities that include headache syndromes other than migraine, other neurological conditions, other pain disorders, sleep disorders, GI disorders, and psychiatric disorders
- In contrast to other epidemiological studies,[8,9] in this real-world patient population, GI comorbidities were more common than those in the psychiatric category

References

1. Smith TR, et al. *Headache.* 2021; 61(8):1214-1226. **2.** TRUDHESA® Package insert. Impel Pharmaceuticals; 2021. **3.** Silberstein SD, et al. *Headache.* 2020;60(1):40-57. **4.** Shrewsbury SB, et al. *Headache.* 2019;59(3):394-409. **5.** Cooper W, et al. *J Aerosol Med Pulm Drug Deliv.* 2022;35(6):321-332. **6.** Buse DC, et al. *Headache.* 2020;DOI:10.1111/head.13966. **7.** Buse DC, et al. *J Neurol Neurosurg Psychiatry.* 2010;81(4):428-432. **8.** Buse DC, et al. *J Headache Pain.* 2020;21(1):23. **9.** Scher AI, et al. *Cephalalgia.* 2019;39(2):296-305.

Disclosures and Acknowledgments

Trupti Gokani reports no disclosures. Linda Nguyen reports serving as an investigator for Allergan and Vanda; a consultant for Abbvie, Ironwood, Alnylam, Eli Lilly, Gemelli, Neurogastrx, Pendulum, Phathom, RosVivo, Salix, and Takeda; and on scientific advisory boards for Gemelli and RosVivo. Robert E. Vann, Judie Gutierrez, Sutapa Ray, and Sheena K. Aurora are full-time employees of Impel Pharmaceuticals and are stockholders in Impel Pharmaceuticals. This research was sponsored by Impel Pharmaceuticals Inc. Editorial support was provided by IMPRINT Science and funded by Impel Pharmaceuticals Inc. IMPEL, POD, and the IMPEL Logo are registered trademarks of Impel Pharmaceuticals Inc.